Pilot trials of the STAR TGC protocol in a cardiac surgery ICU

A.J. Le Compte, S. Penning, K.T. Moorhead, P. Massion, J.C. Preiser, G.M. Shaw, T. Desaive, J.G. Chase

INTRODUCTION

Hyperglycemia has been linked to worsened outcomes, including increased incidence of sepsis, increased ventilator dependence, hospital length of stay and mortality

Tight glycemic control (TGC) has shown particular benefits in cardiac surgery ICU patients. STAR is a model-based TGC protocol accounting for inter- and intra-patient variability with a stochastically derived maximum 5% risk of blood glucose (BG) below 72mg/dL. This abstract describes the first clinical pilot trials of STAR.

CLINICAL TRIALS

Nine cardiac-surgery patients received model-based BG control for 24 hours with BG measured 1-2 hourly. Two-hourly measurements were used when BG was between 110-135mg/dL for 3 hours.

Carbohydrate intake (all sources) was monitored, but not changed from clinical settings except to prevent potential low BG. Insulin infusion rates were limited (6U/hour maximum), with limited increases based on current infusion rate (0.5-2.0U/hour). Approval was granted by the Ethics Committee of the Medical Faculty of the University of Liege (Liege, Belgium).

MODELS AND METHODS

A metabolic computer system model, clinically validated in adult intensive care patients and virtual trials using neonatal data, is used in conjunction with a stochastic model of insulin sensitivity variability to provide tight control:

1. Previous and current blood glucose measurements are used to determine a patient-specific real-time insulin sensitivity estimate.
2. The stochastic model provides a distribution of possible insulin sensitivity parameter values for the next 1-2 hours.
3. Insulin rate required to achieve BG target of 125 mg/dL is computed with a bisection method.
4. BG outcome predictions are calculated for the 5th, 25th, 75th and 95th percentile SI values over the next 1-2 hours.
5. Predicted outcome BG range is checked to ensure the lowest possible BG is not < 72 mg/dL. If lowest BG is < 72 mg/dL, the insulin rate is reduced to ensure a guaranteed maximum risk of 5% of BG < 72 mg/dL for safety.

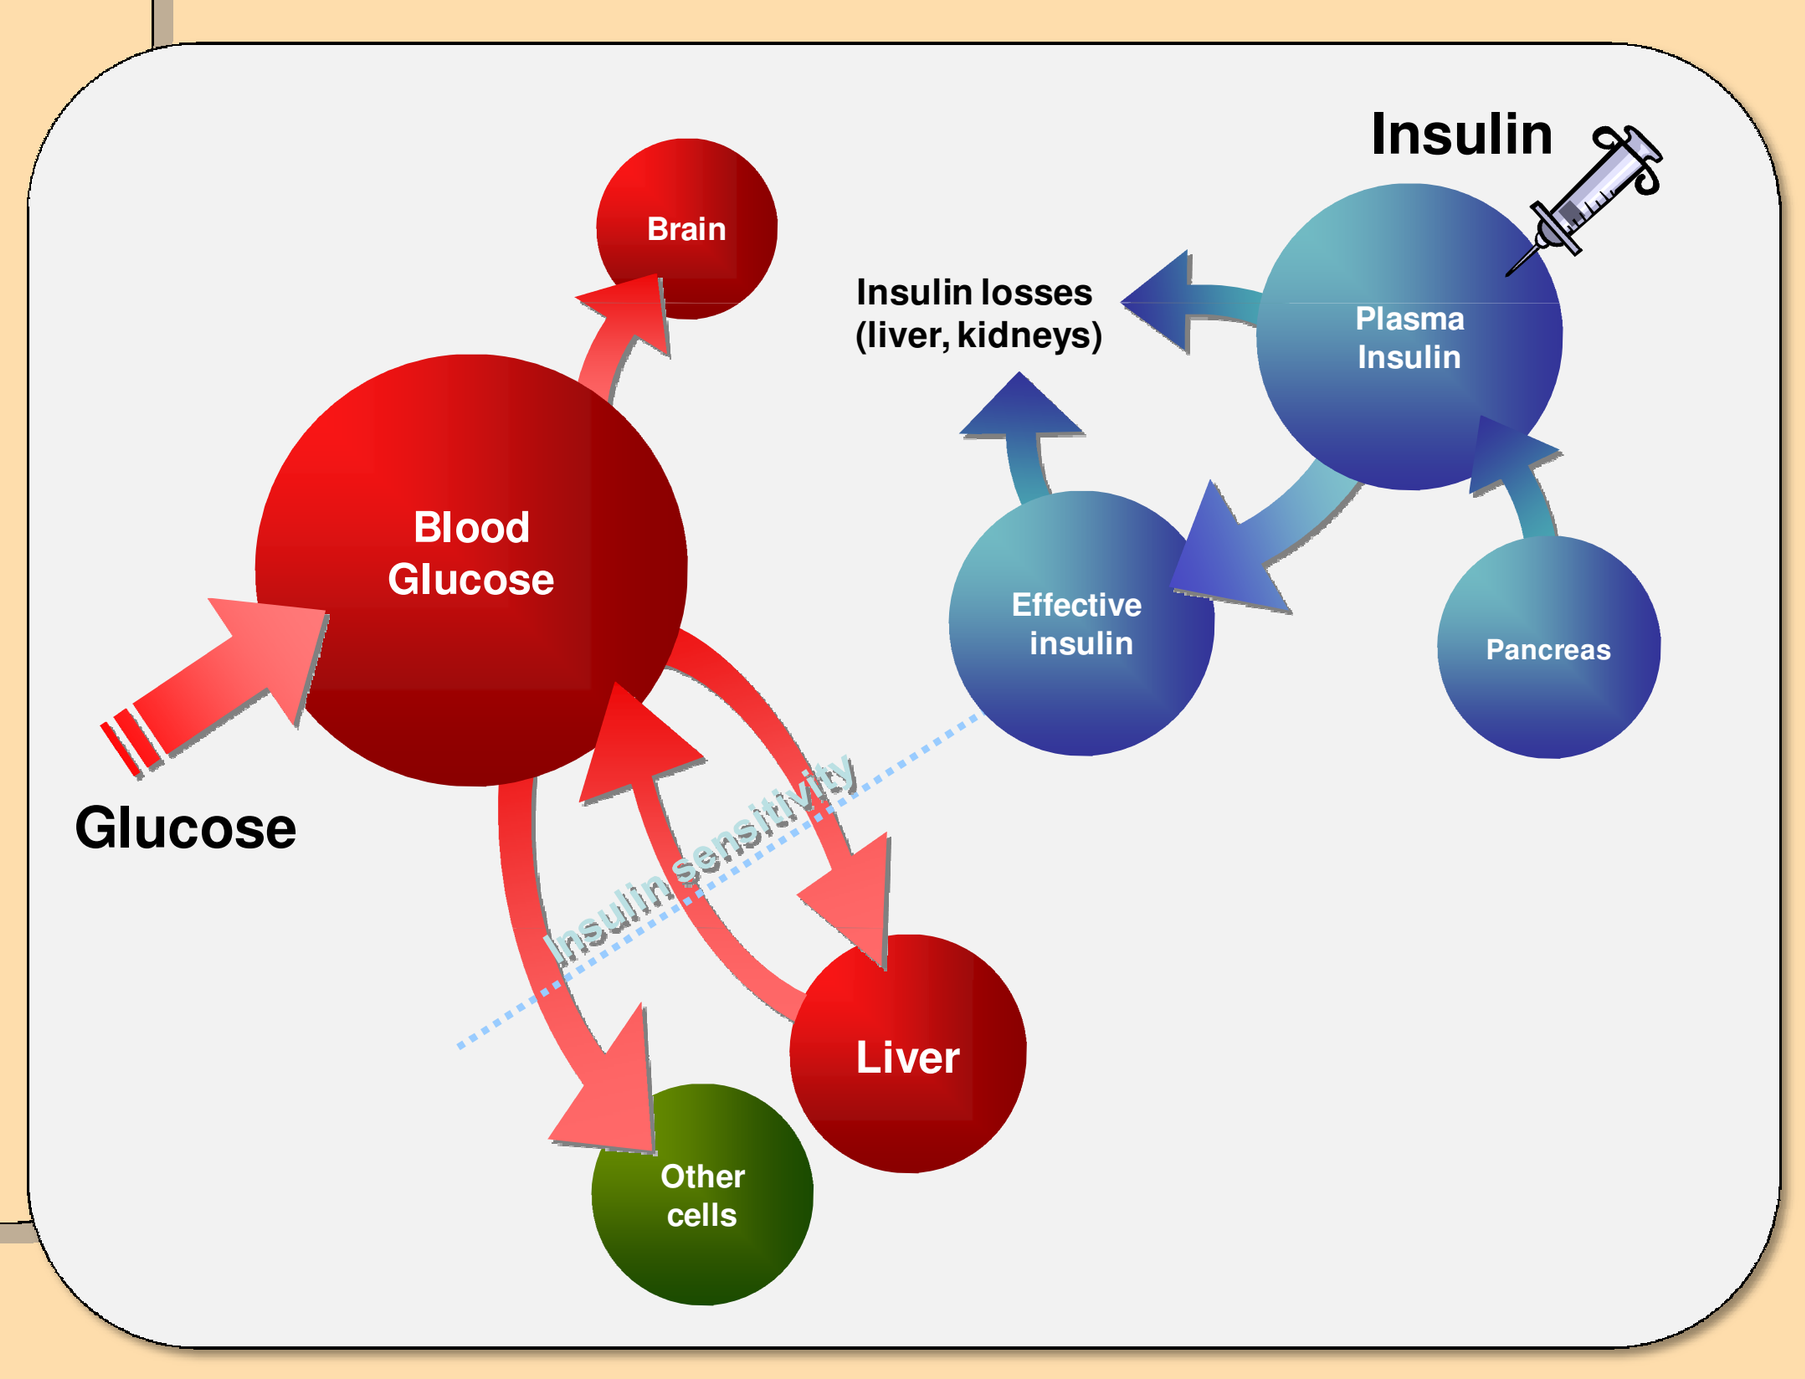
Insulin
Brain
Insulin losses (liver, kidneys)
Plasma Insulin
Blood Glucose
Effective insulin
Pancreas
Glucose
Insulin sensitivity
Liver
Other cells

Figure 1: Schematic representation of glucose-insulin model (left) and model equations (below). The insulin sensitivity parameter SI is fitted to BG, insulin and nutrition data, and forecasted using stochastic modeling methods.

$$\dot{G} = -p_G.G - \mathrm{SI}(t)*G.\frac{Q}{1+\alpha_G Q} + \frac{P(t) + (P_{END}*m_{body}) - (CNS*m_{brain})}{(V_{G,frac}(t)*m_{body})}$$

$$\dot{Q} = -kQ + kI$$

$$\dot{I} = -\frac{nI}{1+\alpha_I I} + \frac{u_{ex}(t)}{(V_{I,frac}*m_{body})} + e^{-(k_I \frac{u_{ex}(t)}{V_i})} I_B$$

RESULTS

Overall trial results are presented in Table 1. There were 205 BG measurements taken during 215 hours of control. No hypoglycemia (BG < 40 mg/dL) occurred during these clinical pilot trials. The minimum BG was 63mg/dL. Average model prediction error was 13.9 mg/dlL (10.5%), with larger errors due to small meals and other clinical events.

Carbohydrate intake varied widely between patients (4.2 [2.0 - 11.1] g/hour), and zero insulin rates were determined by the control system for 25% of interventions (Figure 2).

Insulin sensitivity in trial patients was significantly more variable than general medical ICU patients (Figure 3), but similar to recent post-surgery cardiac patients from other cohorts. The trial patients also exhibited relatively high sensitivity to insulin, contributing to the potential for increased variability in glycemic outcome.

Table 1: Overall STAR pilot clinical trial results. Reduction of hyperglycemia to median BG levels < 140 mg/dL was achieved with no hypoglycemic events.

Whole cohort statistics		Per-patient statistics (presented as median [IQR])	
Number of patients:	9	Hours of control:	24.0 [23.0 - 24.3]
Total hours:	215 hours	Num BG measurements:	24.0 [22.0 - 24.0]
Num BG measurements:	205	Initial BG (mg/dL):	160.0 [151.5 - 172.0]
BG median [IQR] (mg/dL):	136.0 [122.5 - 158.0]	BG median (mg/dL)	138.5 [130.6 - 146.0]
% BG within 80-110 mg/dL	9.8	Median %BG within 80-110 mg/dL [IQR]:	8.3 [4.5 - 9.9]
% BG within 110-140mg/dL	47.3	Median %BG within 110-140 mg/dL [IQR]:	52.6 [33.3 - 55.5]
% BG within 140-180mg/dL	37.6	Median %BG within 140-180 mg/dL [IQR]:	40.9 [29.5 – 41.7]
% BG > 180 mg/dL	7.8	Median %BG > 180 mg/dL [IQR]:	8.3 [0.0 - 14.4]
% BG < 80 mg/dL	0.5	Time to < 140 mg/dL (hours):	2.1 [2.0 - 4.0]
% BG < 72 mg/dL	0.5	% patients to < 140 mg/dL:	100
% BG < 40 mg/dL	0		
Num patients < 40 mg/dL	0	Max insulin rate [IQR] (U/hr):	6.0 [4.7 - 6.0]
Median insulin rate [IQR] (U/hr):	1.5 [0.5 - 3.4]	Median insulin rate [IQR] (U/hr):	1.3 [0.9 - 2.4]
Median glucose rate [IQR] (g/hour):	7.4 [2.0 - 11.2]	Median dextrose rate [IQR] (g/hour):	4.2 [2.0 - 11.1]

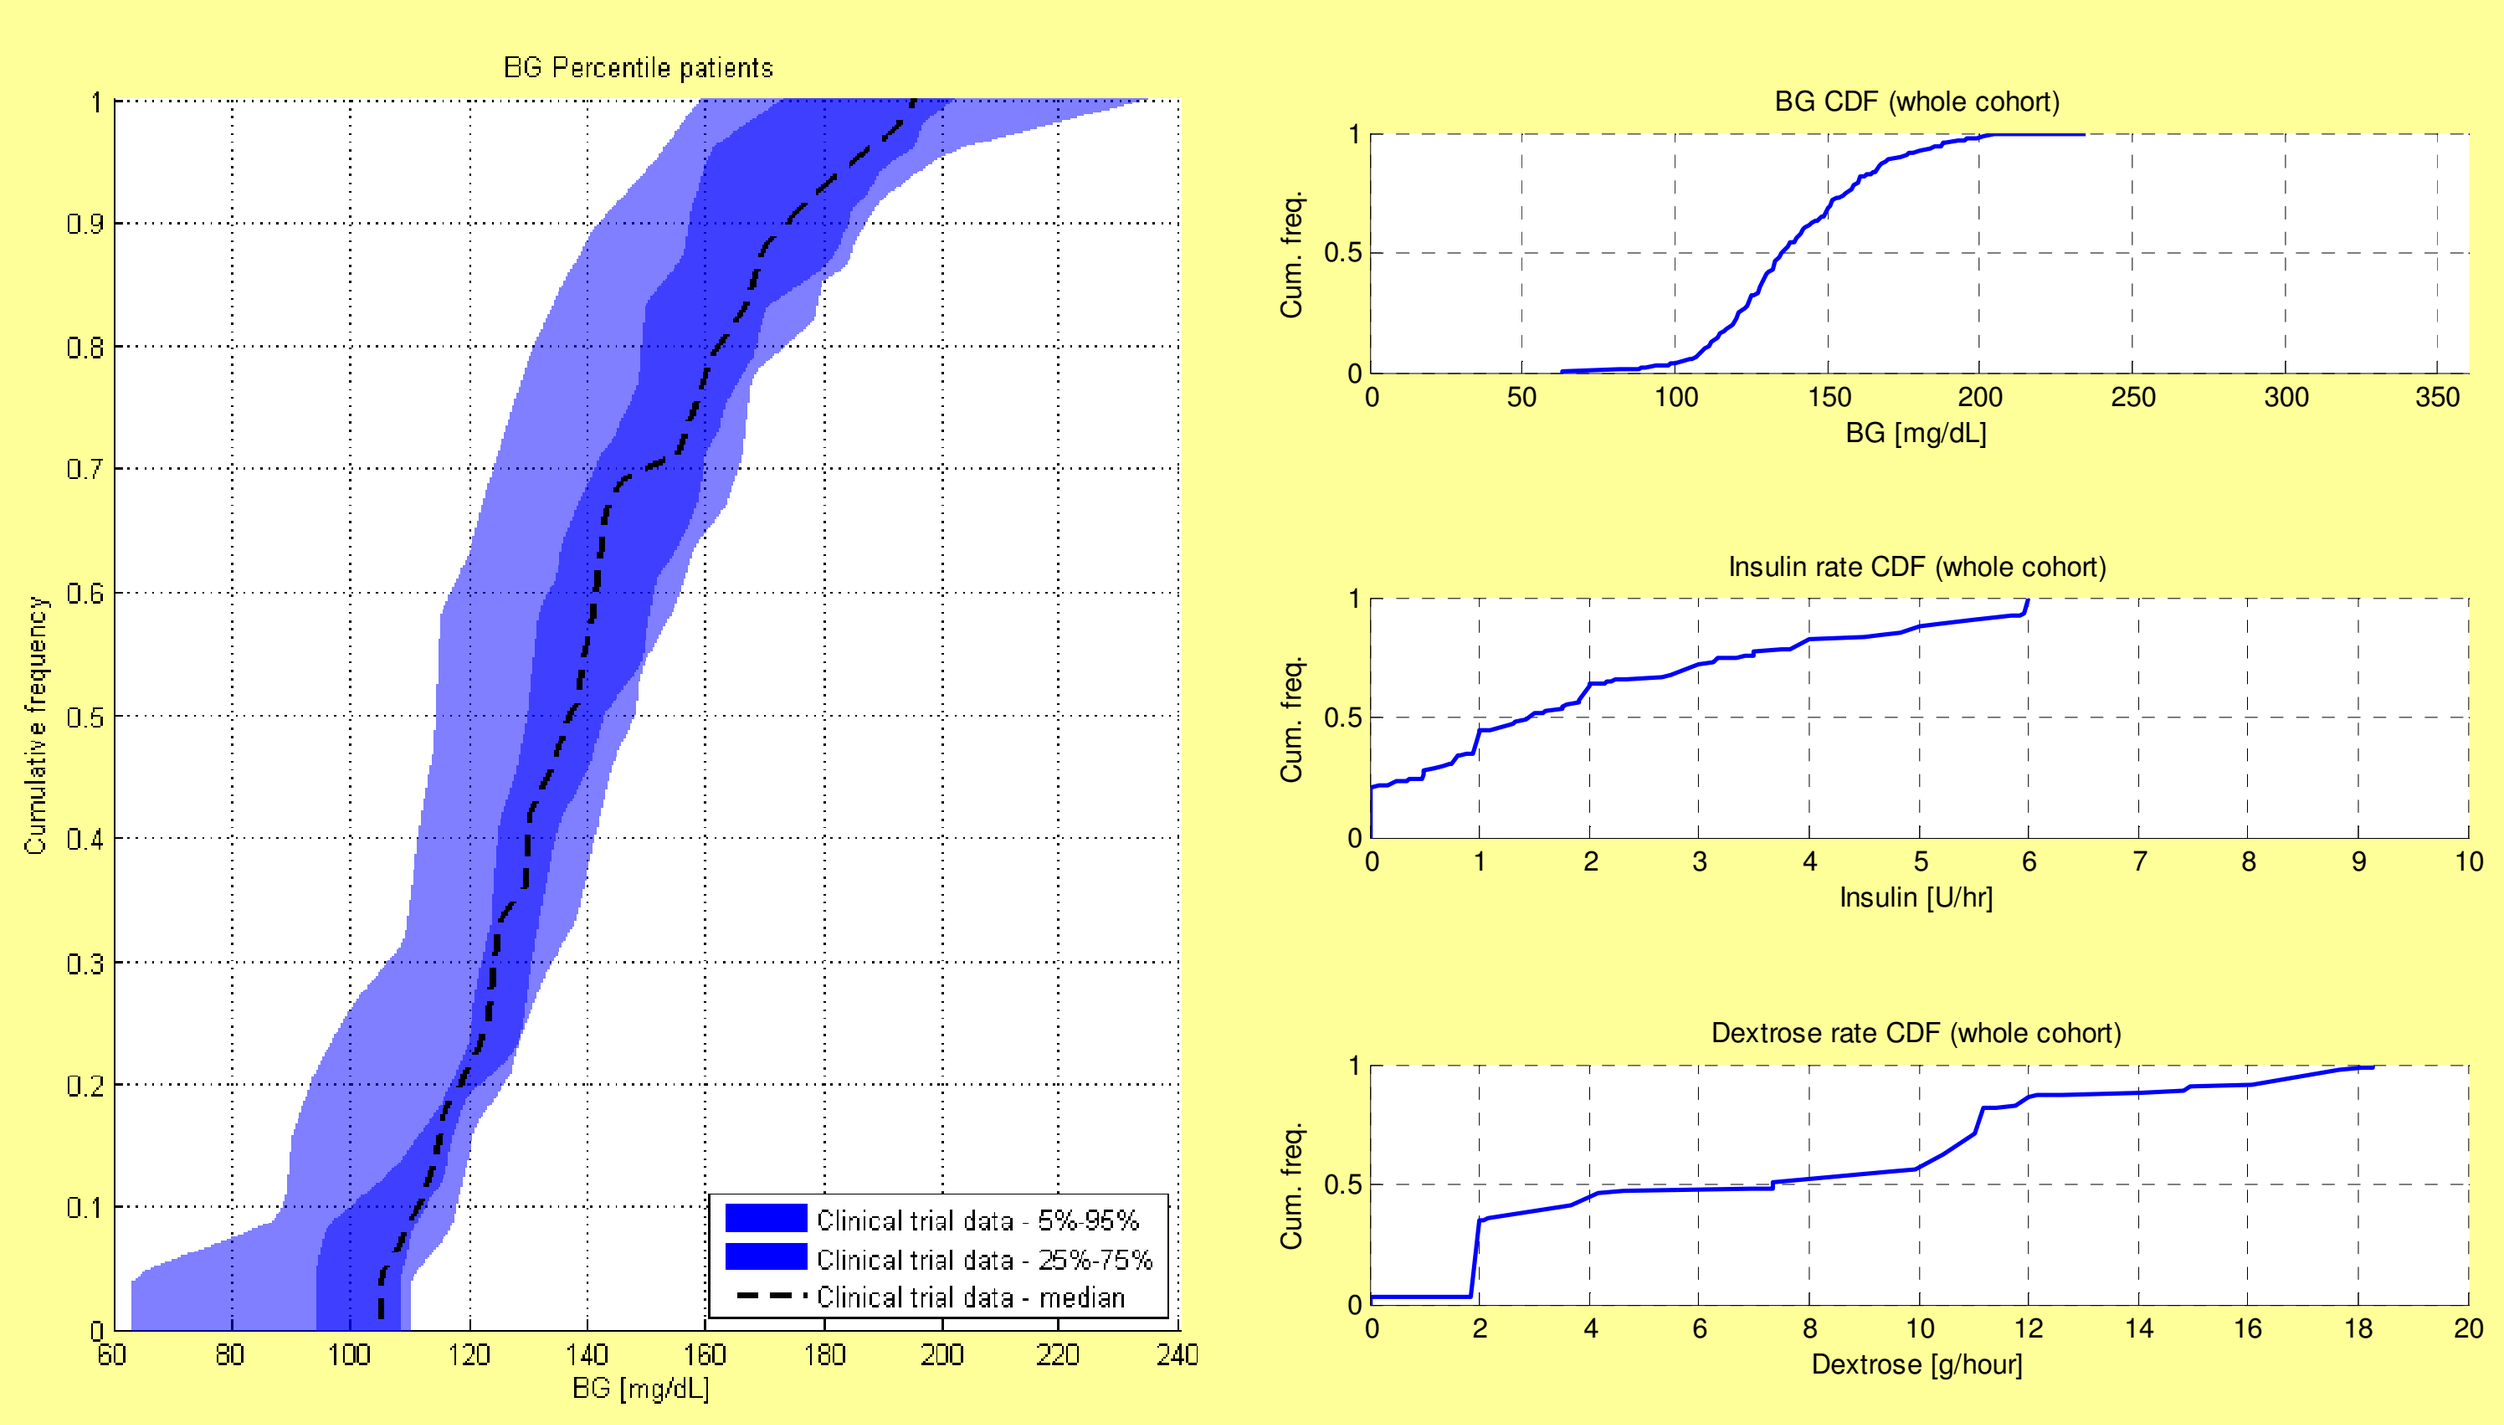
BG Percentile patients
Cumulative frequency
BG [mg/dL]
Clinical trial data - 5%-95%
Clinical trial data - 25%-75%
Clinical trial data - median
BG CDF (whole cohort)
Cum. freq.
BG [mg/dL]
Insulin rate CDF (whole cohort)
Insulin [U/hr]
Dextrose rate CDF (whole cohort)
Dextrose [g/hour]

Figure 2: Cumulative density functions of blood glucose concentration and insulin/dextrose inputs. The left panel displays 5th, 25th, 50th, 75th and 95th percentiles of per-patient BG distributions. The right panel displays distributions of measured BG (top), insulin (middle) and dextrose (bottom).

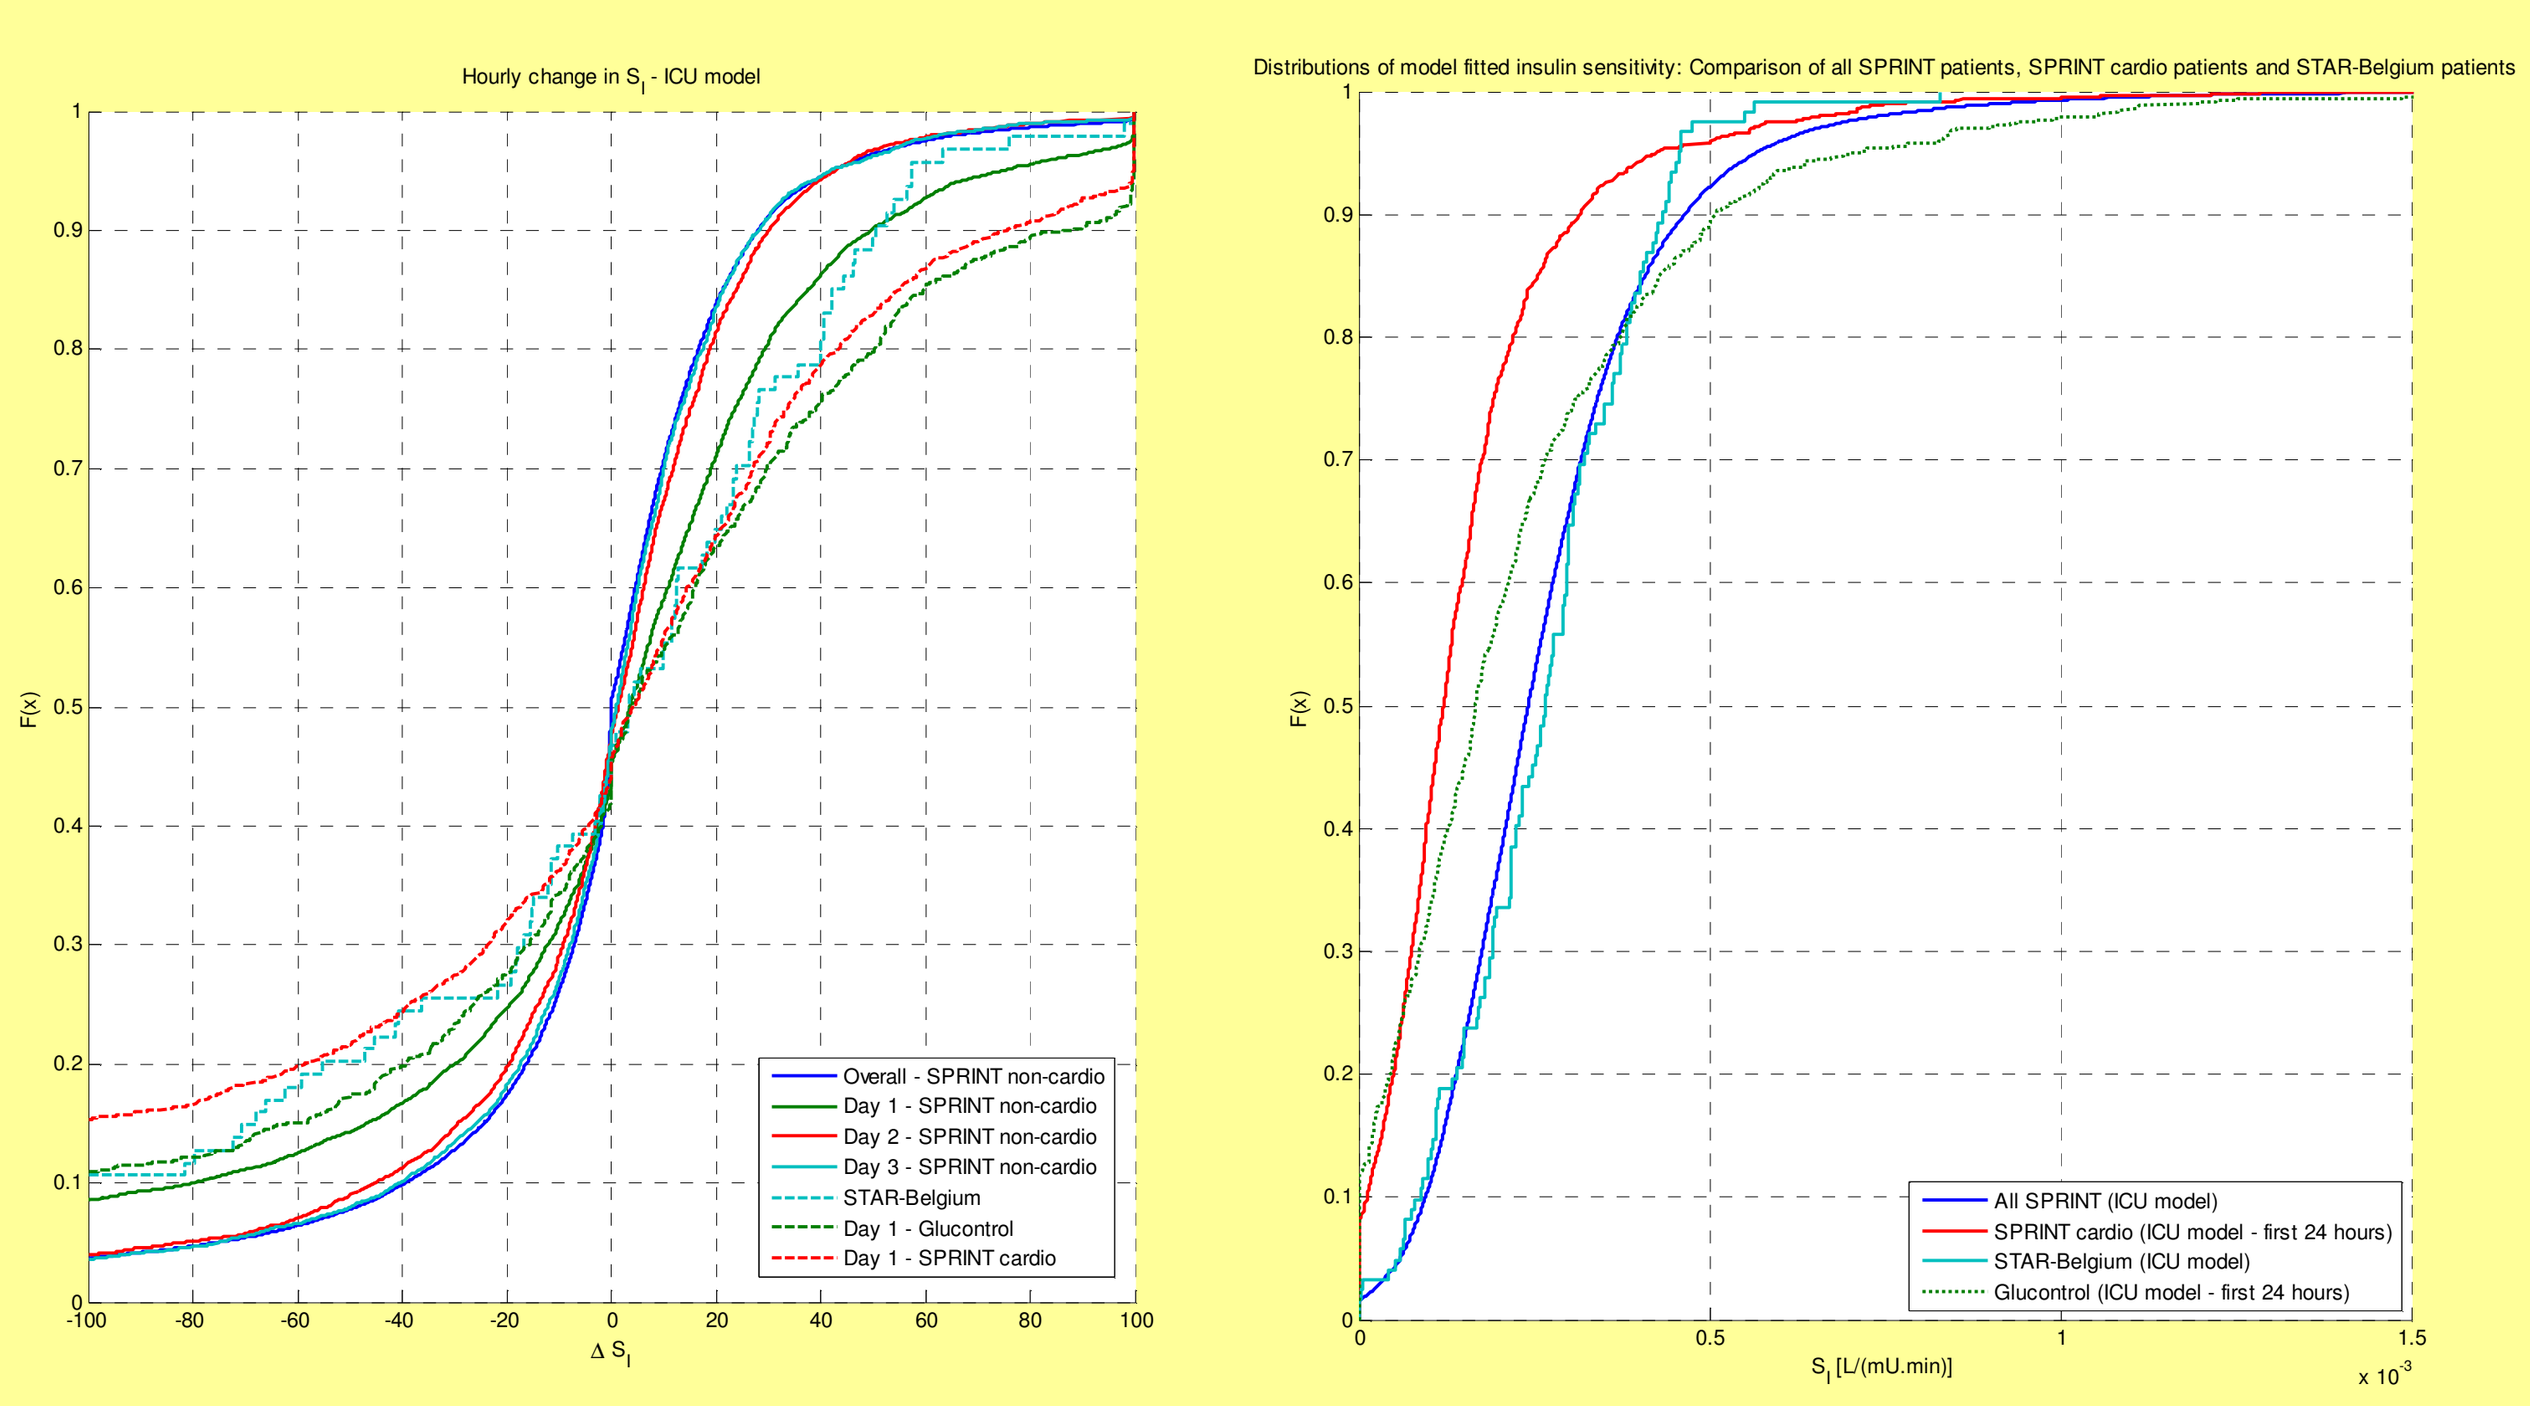
Distributions of model fitted insulin sensitivity: Comparison of all SPRINT patients, SPRINT cardio patients and STAR-Belgium patients
F(x)
Overall - SPRINT non-cardio
Day 1 - SPRINT non-cardio
Day 2 - SPRINT non-cardio
Day 3 - SPRINT non-cardio
STAR-Belgium
Day 1 - Glucontrol
Day 1 - SPRINT cardio
All SPRINT (ICU model)
SPRINT cardio (ICU model - first 24 hours)
STAR-Belgium (ICU model)
Glucontrol (ICU model - first 24 hours)

Figure 3: Cumulative distribution functions of hourly % change in insulin sensitivity (left panel) and absolute insulin sensitivity distribution (right panel). The STAR patients exhibited similar trends in insulin sensitivity variability compared to cardiac surgery cohorts on the first day of BG control, and substantially higher overall sensitivity to insulin compared to cardiac-surgery patients from a general ICU.

Conclusion:

STAR effectively controlled all patients towards target. Observed patient variability in response to insulin and thus prediction errors were higher than expected, likely due to the recent insult of cardiac surgery and their immediate recovery. STAR effectively managed this enhanced variability with no hypoglycemia, and the high density BG data allows comparison of variability between surgical and medical ICU patients.

Université de Liège
UC BIOMEDICAL ENGINEERING
UC UNIVERSITY OF CANTERBURY Te Whare Wānanga o Waitaha CHRISTCHURCH NEW ZEALAND