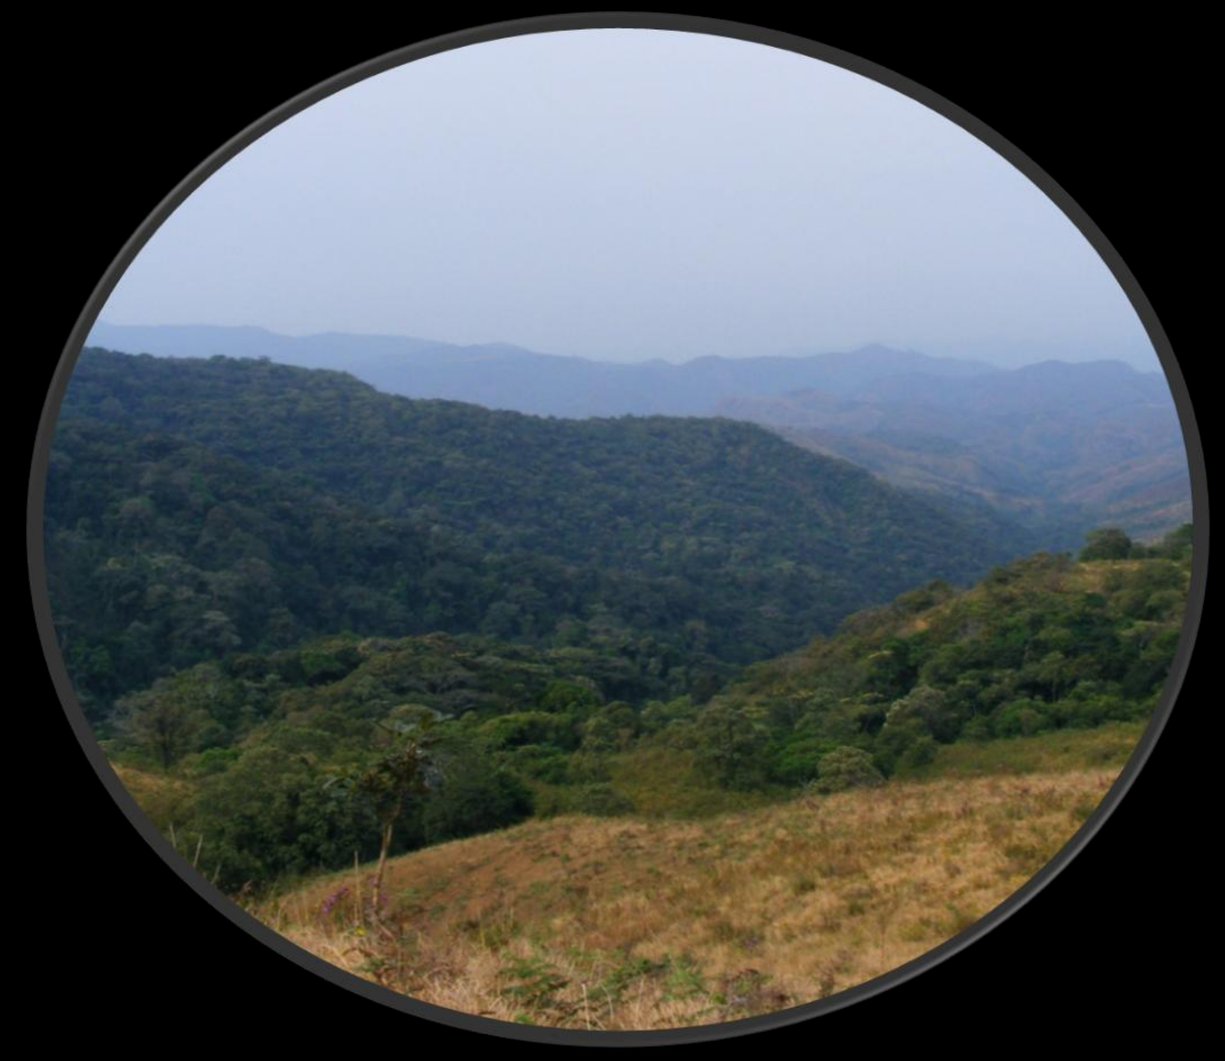

The Effects Of Forest Edges On Dung Beetle Communities In A Tropical Montane Forest

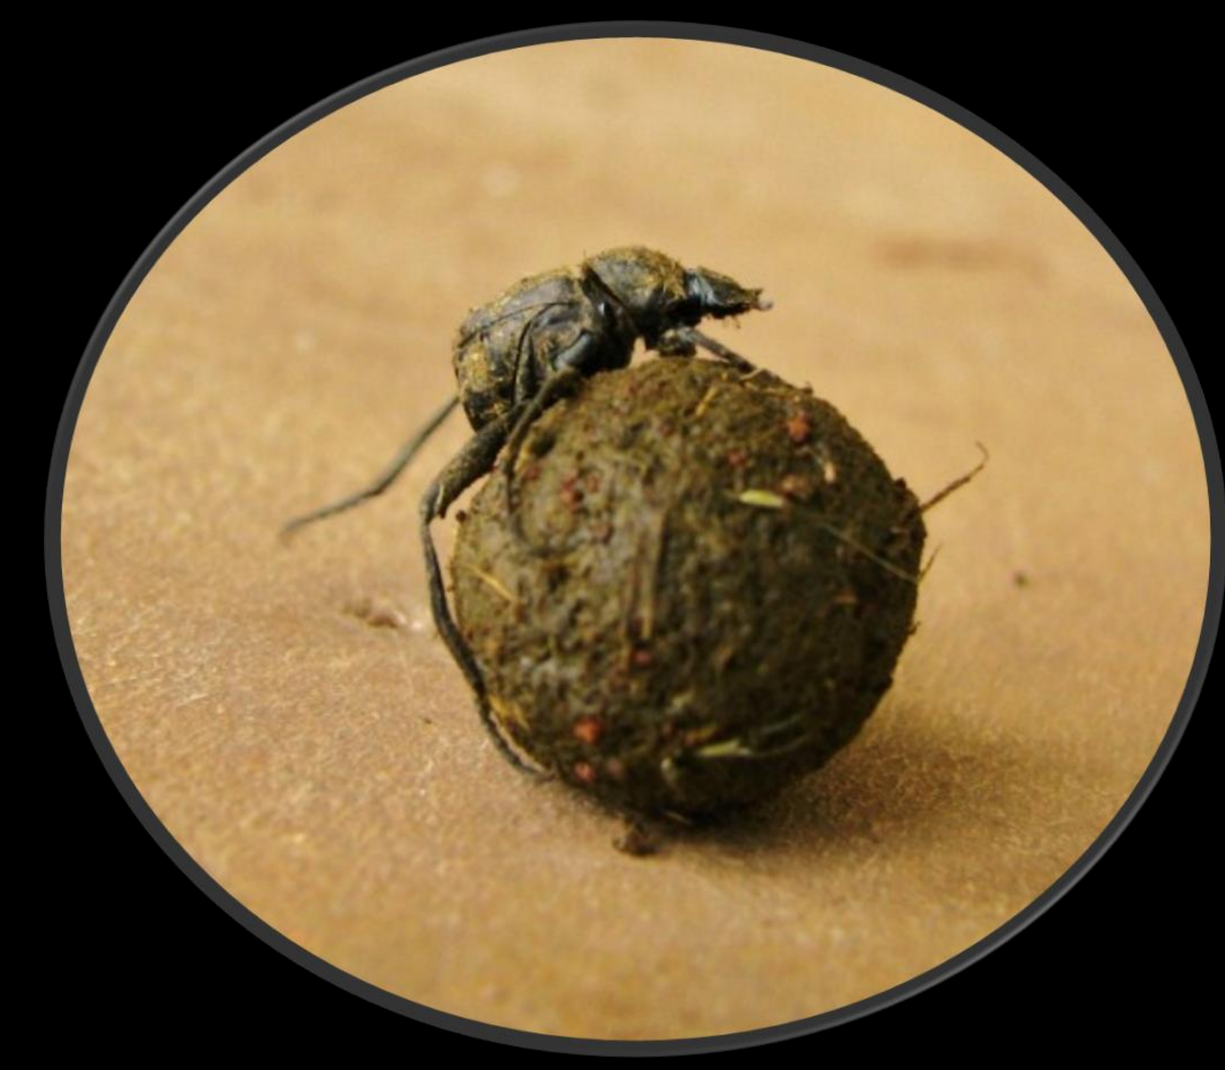

Andrew D. Barnes[1], Rowan M. Emberson[2], Hazel M. Chapman[1], Raphael K. Didham[1,3]

[1]School of Biological Sciences, University of Canterbury, Private Bag 4800, Christchurch, New Zealand
[2]Department of Entomology and Animal Ecology, Lincoln University, P.O. Box 84, Lincoln, New Zealand
[3]School of Animal Biology, University of Western Australia, 35 Stirling Highway, Crawly WA 6009, Australia

Living On The Edge

Land use has been implicated as the largest global driver of biodiversity loss, largely due to associated habitat loss and fragmentation. The resulting production of habitat edges have pervasive impacts on the distribution and persistence of invertebrates[1]. Land use change is of particular concern in African tropical montane forests as populations are increasing dramatically throughout these areas. Therefore , this study focuses on the impacts of livestock and fire on forest edges around a unique Afromontane forest in Nigeria

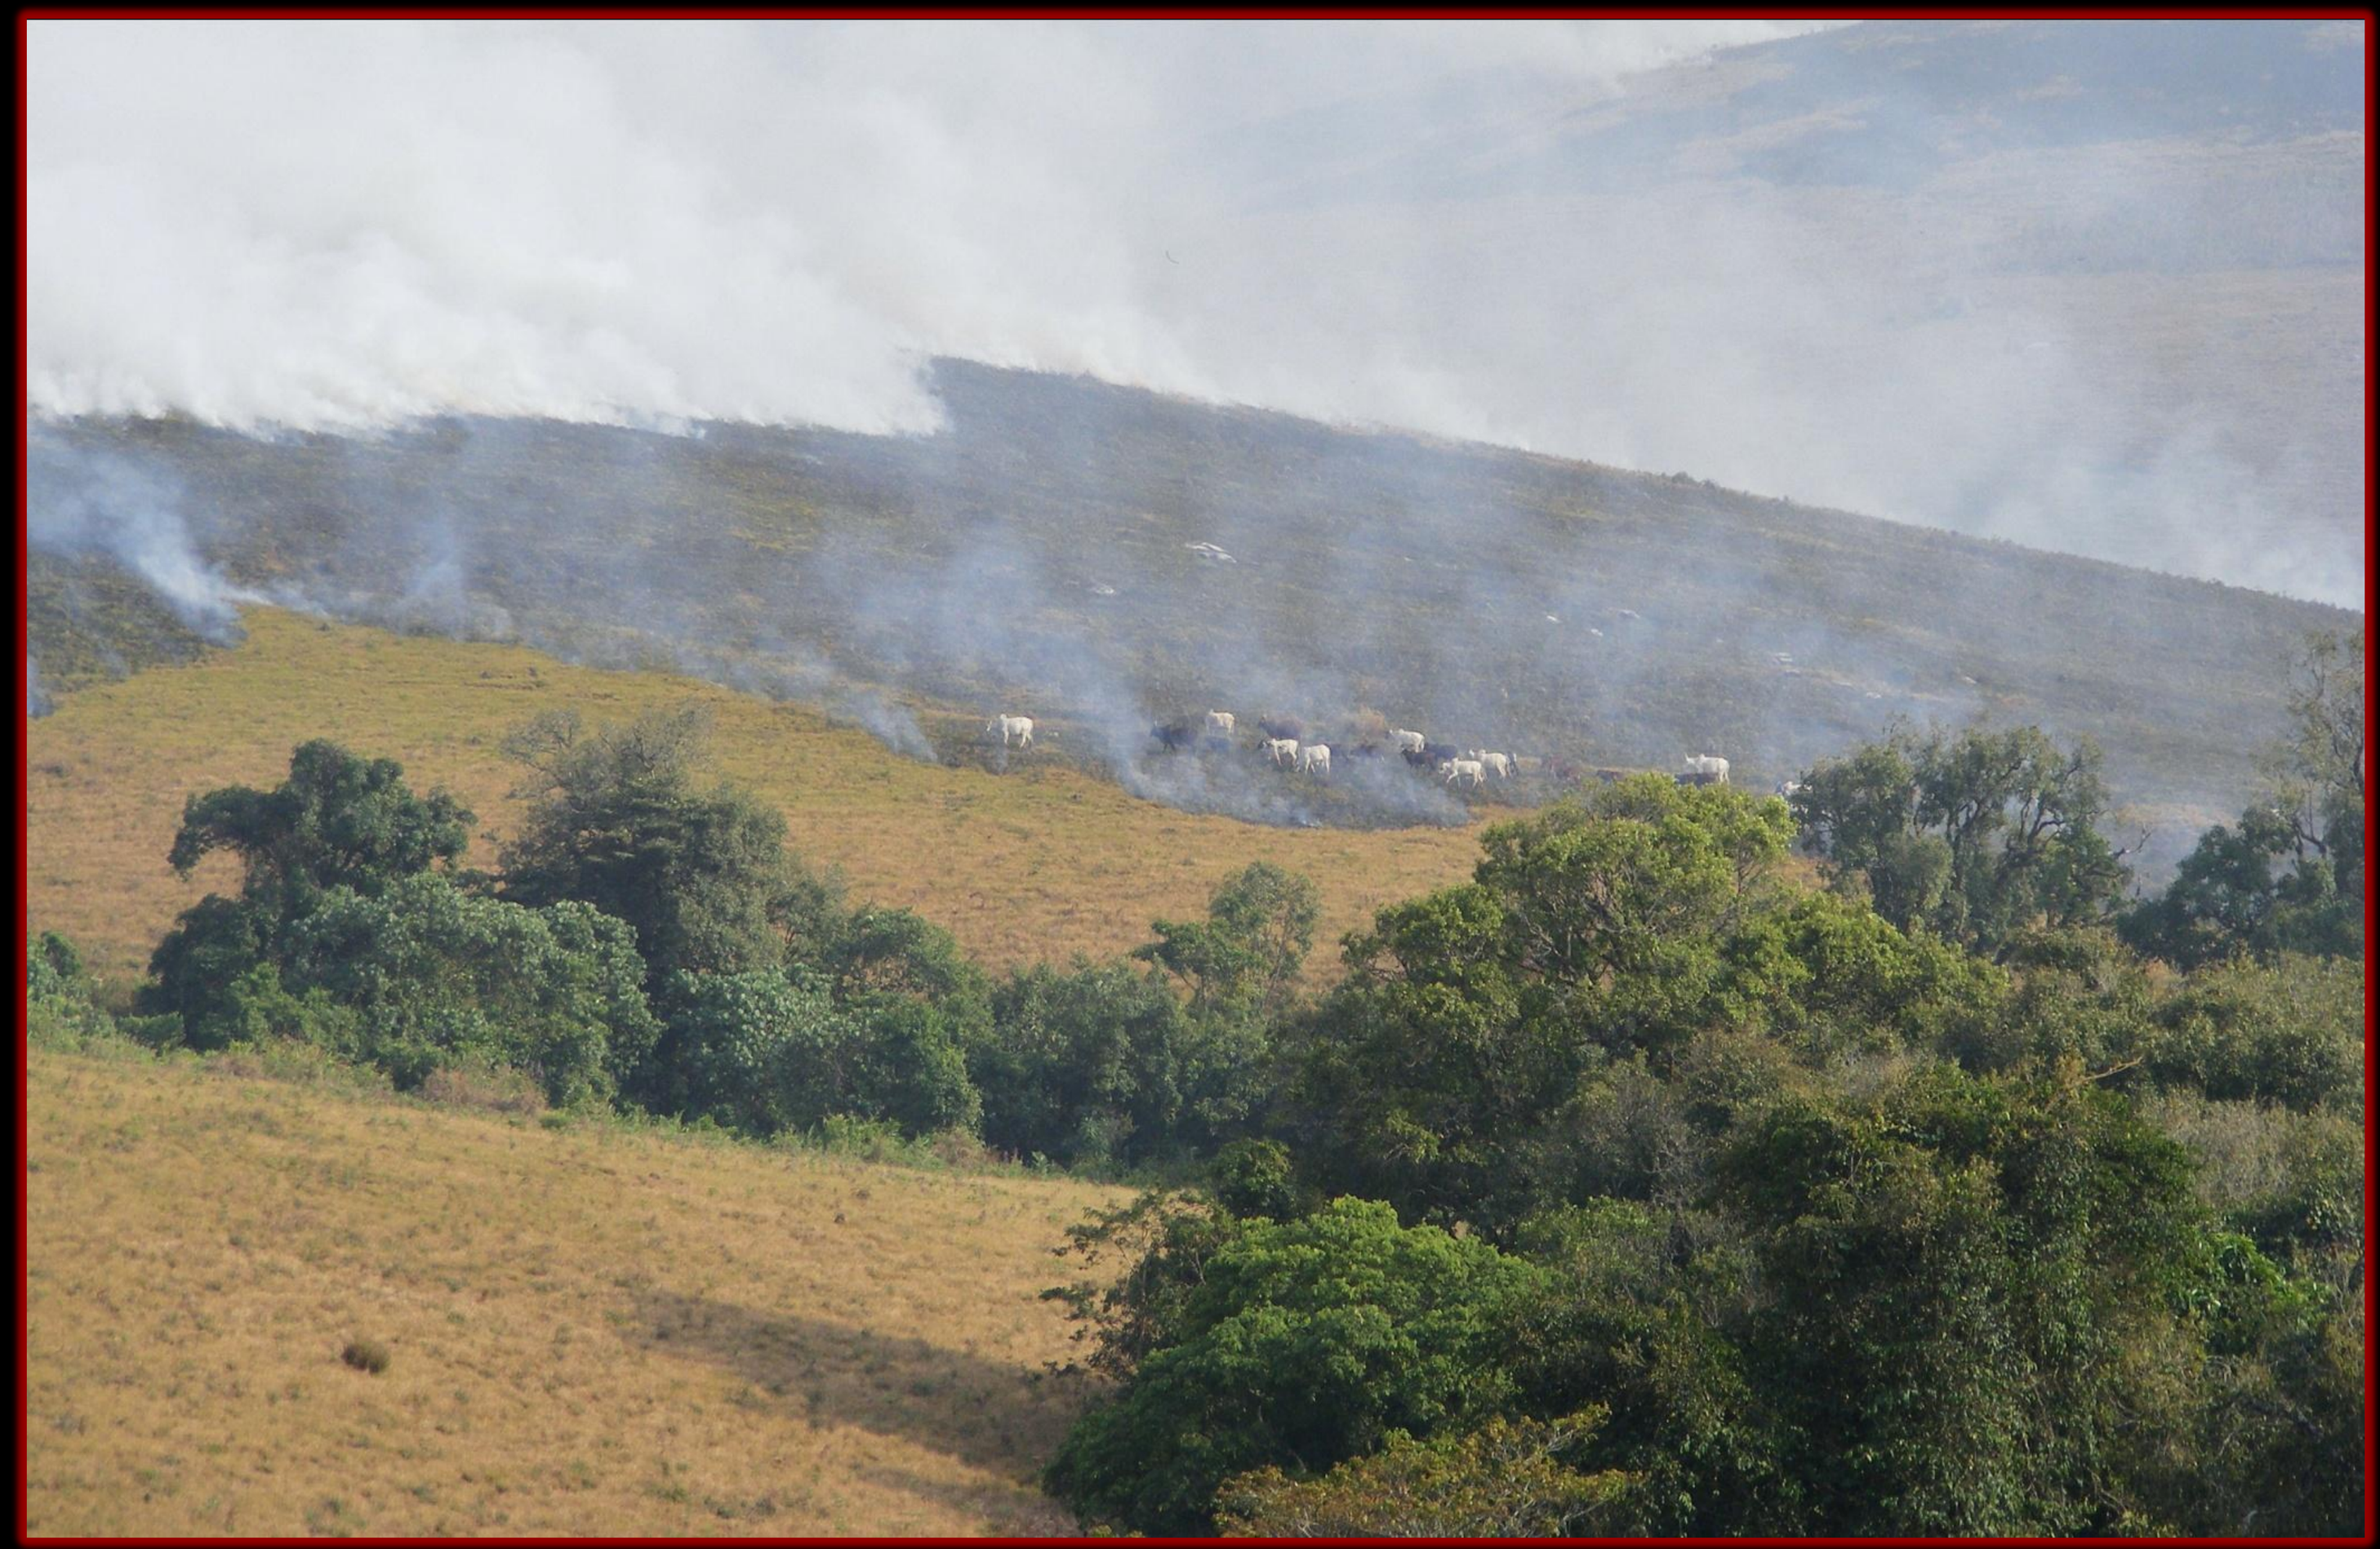

Diversity & Abundance Distributions

Total abundance of dung beetles will be measured per trap at each distance from the edge. Additionally, species diversity and richness will be measured across the edge gradient. This will give an indication of the impact of edge effects and the interactions with matrix/edge condition on the composition of dung beetle communities. This data can then be used to analyse correlations with ecosystem function.

Preliminary findings shown below, present a clear trend in overall abundances of dung beetles in response to distance from the edge. Values are mean abundance ± SE. Additionally, there is a striking difference in abundance distributions between protected (A) and non-protected (B) edges.

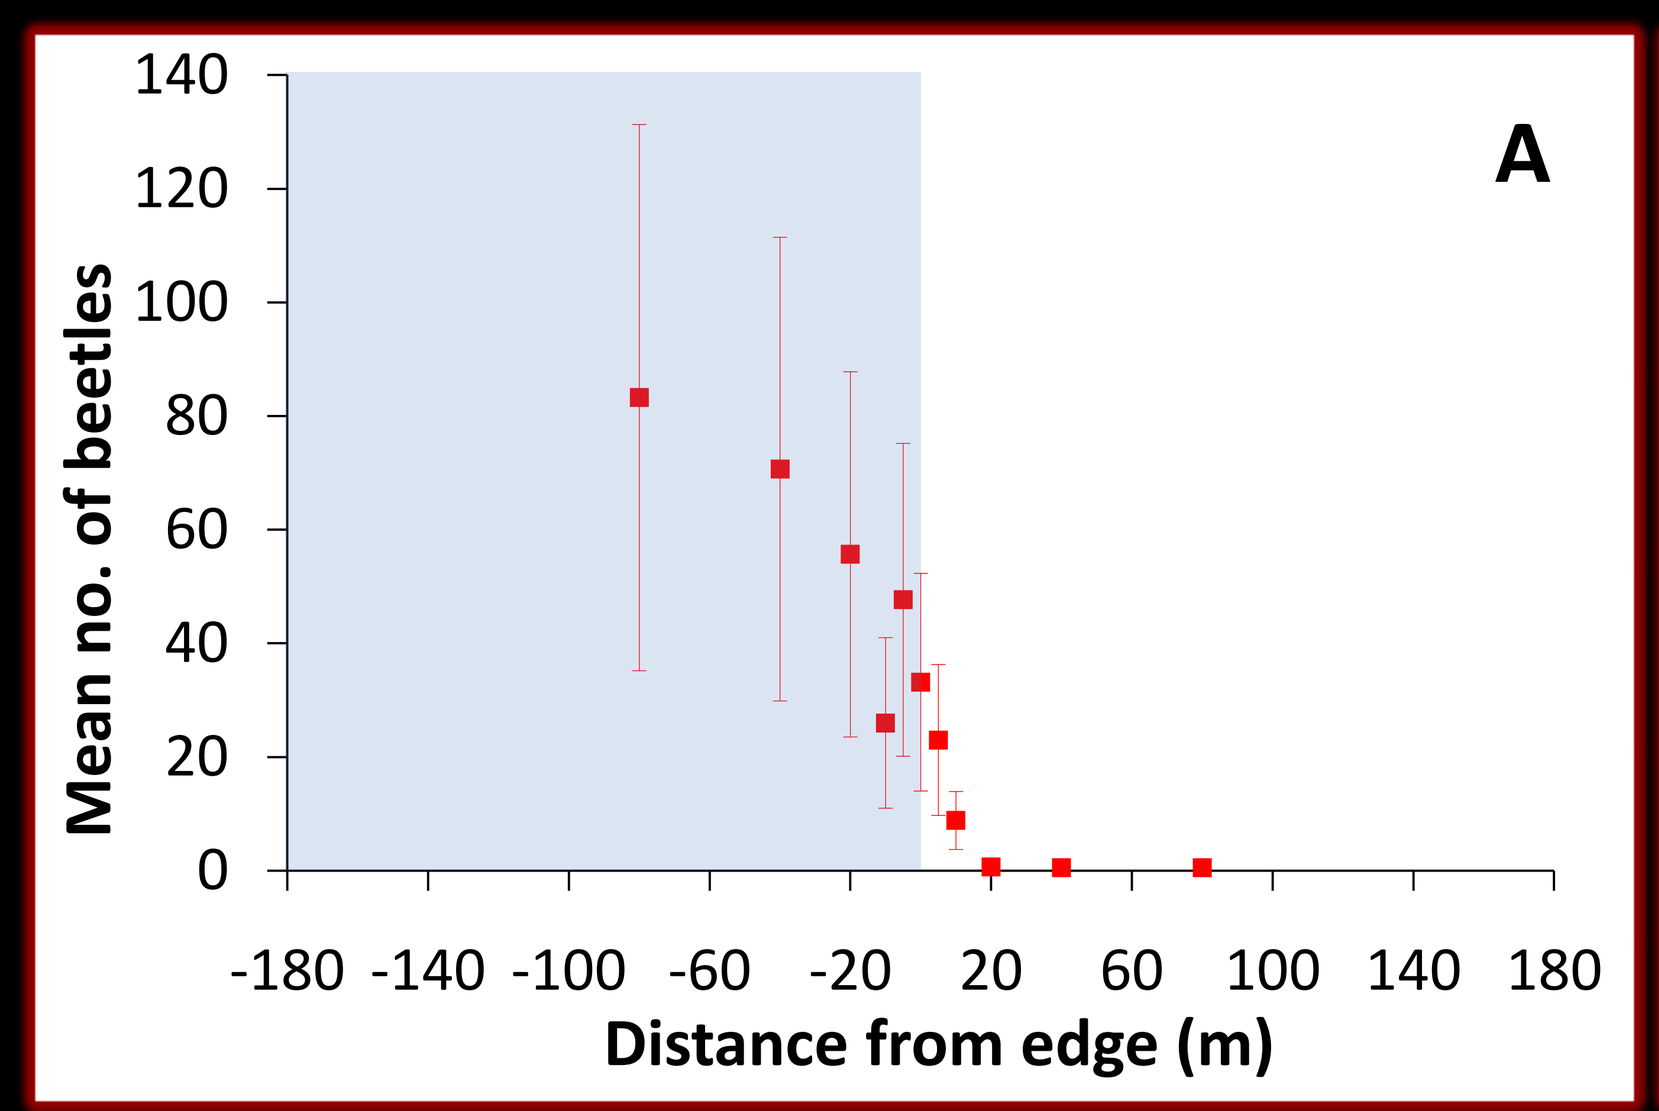
A
Mean no. of beetles
0
20
40
60
80
100
120
140
-180 -140 -100 -60 -20 20 60 100 140 180
Distance from edge (m)

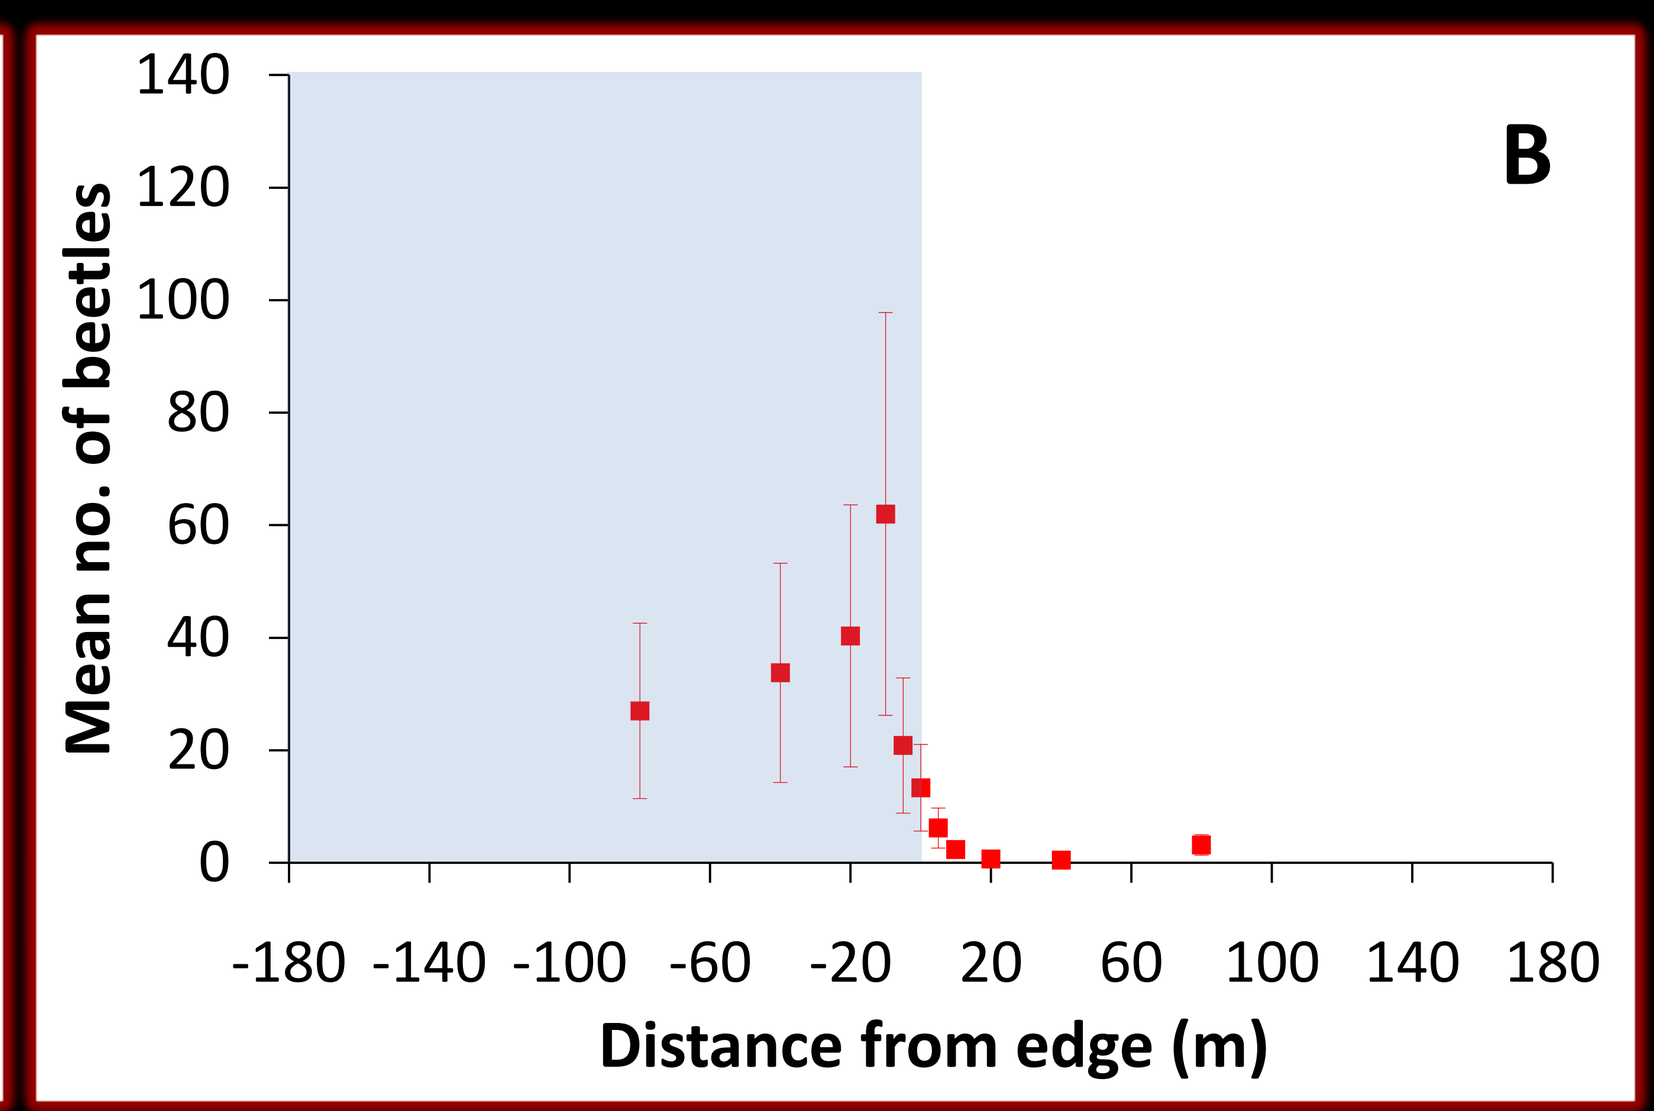
B
Mean no. of beetles
0
20
40
60
80
100
120
140
-180 -140 -100 -60 -20 20 60 100 140 180
Distance from edge (m)

Ecosystem Function

Dung beetles are extremely important as they perform major ecosystem functions by burying dung. This increases rates of decomposition and thus nutrient cycling is enhanced. Additionally, seeds within the dung are secondarily dispersed, reducing seed predation and facilitating germination. Dung removal rates were measured across the edge to test for the effects of habitat edges on ecosystem functioning

Rates of dung removal were measured at the same sites and distances used for trapping. 40gm pieces of dung were placed on the ground for 24 hours, after which total dry weight loss was measured. Preliminary data for removal rates between protected (A) and non-protected (B) edges are shown below . Values are mean percentages of dung removed ± SE.

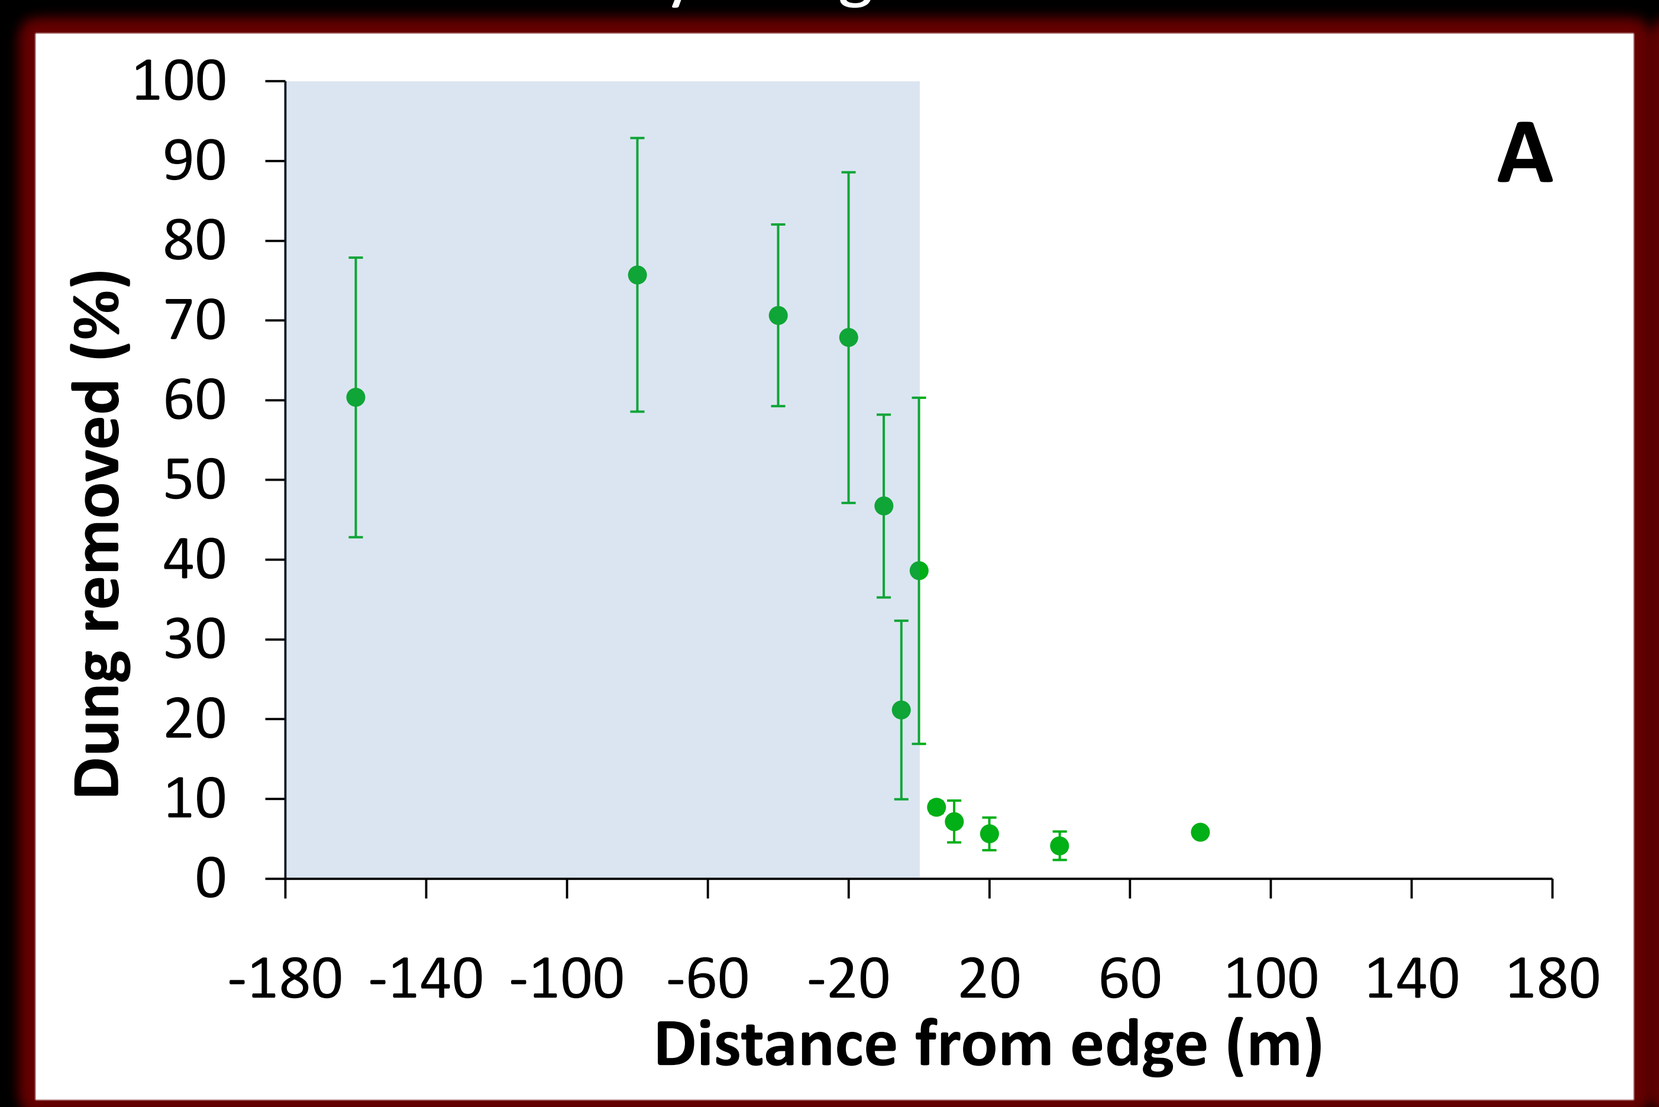
A
Dung removed (%)
0
10
20
30
40
50
60
70
80
90
100
-180 -140 -100 -60 -20 20 60 100 140 180
Distance from edge (m)

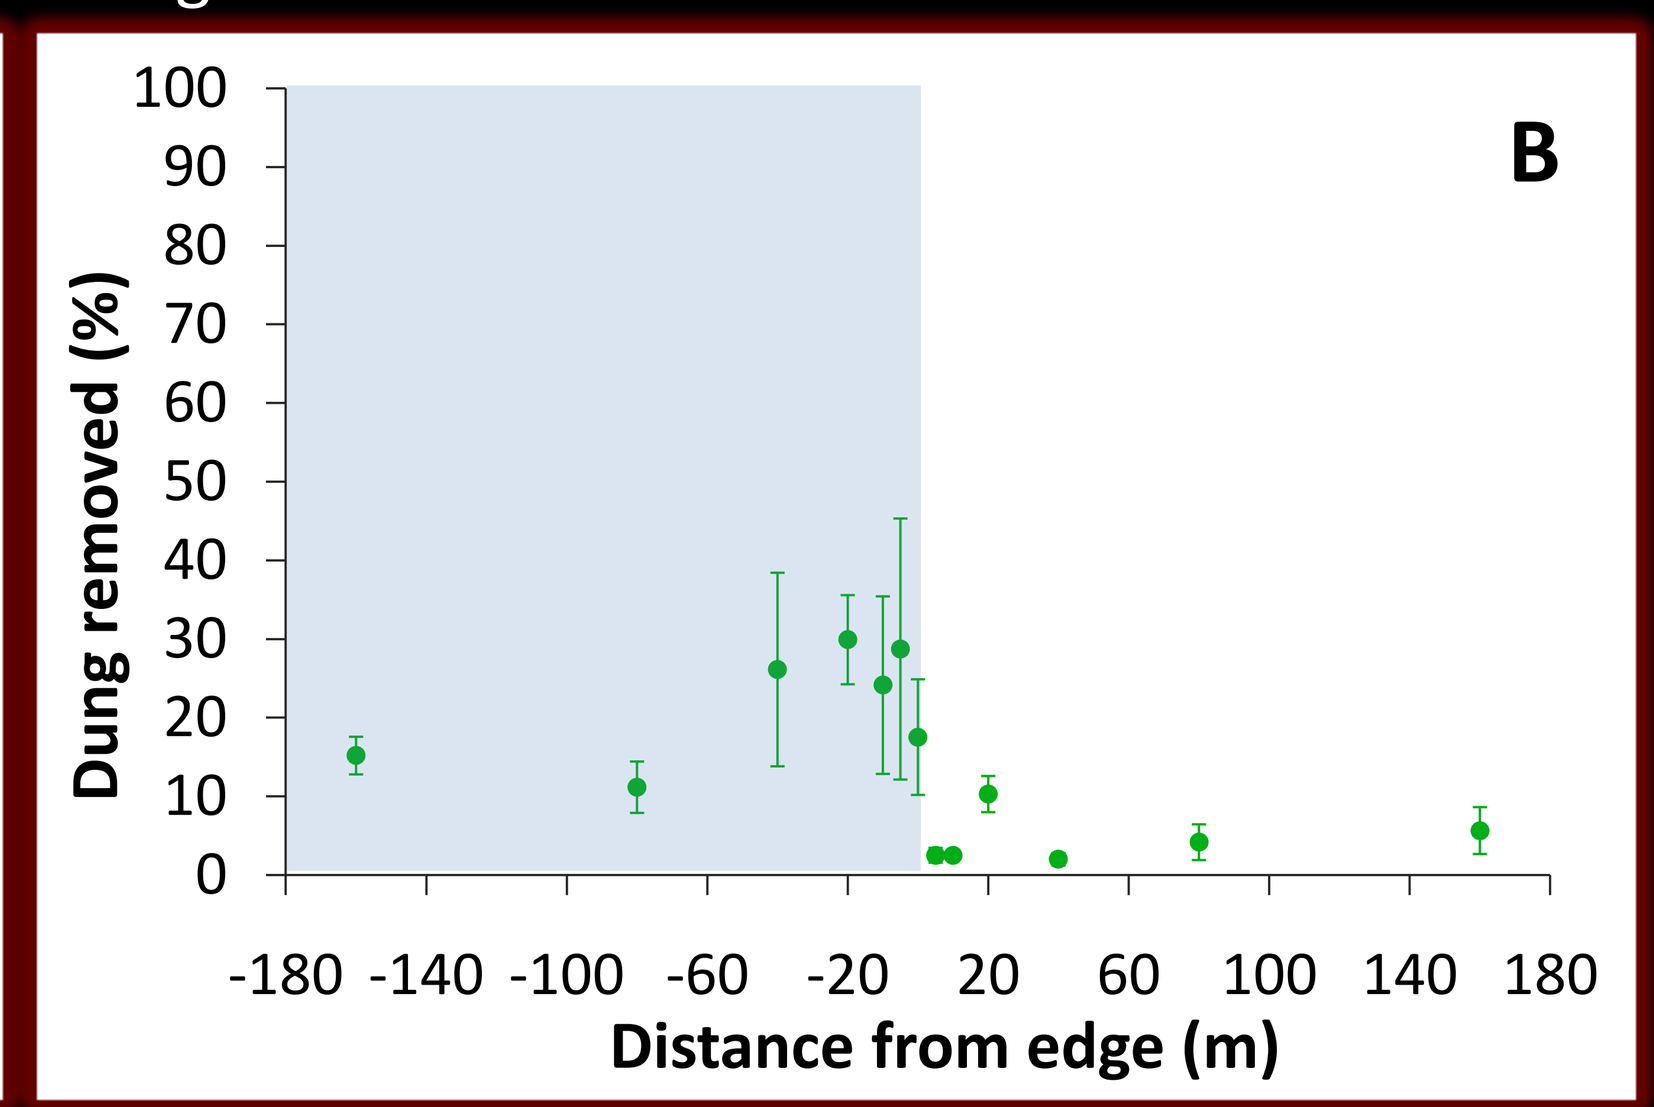
B
Dung removed (%)
0
10
20
30
40
50
60
70
80
90
100
-180 -140 -100 -60 -20 20 60 100 140 180
Distance from edge (m)

Study Site & Design

The effects of anthropogenically created edges on dung beetle community structure has, thus far, not been investigated across a continuous edge gradient. This presents an important gap in our knowledge of the potential effects of surrounding land use on the intensification of edge effects and resulting impacts on dung beetle communities. Dung Beetle communities were sampled at forest edges, both protected (A) and unprotected (B) from intensive livestock grazing in a severely fragmented montane forest landscape surrounding the Ngel Nyaki forest reserve in Nigeria.

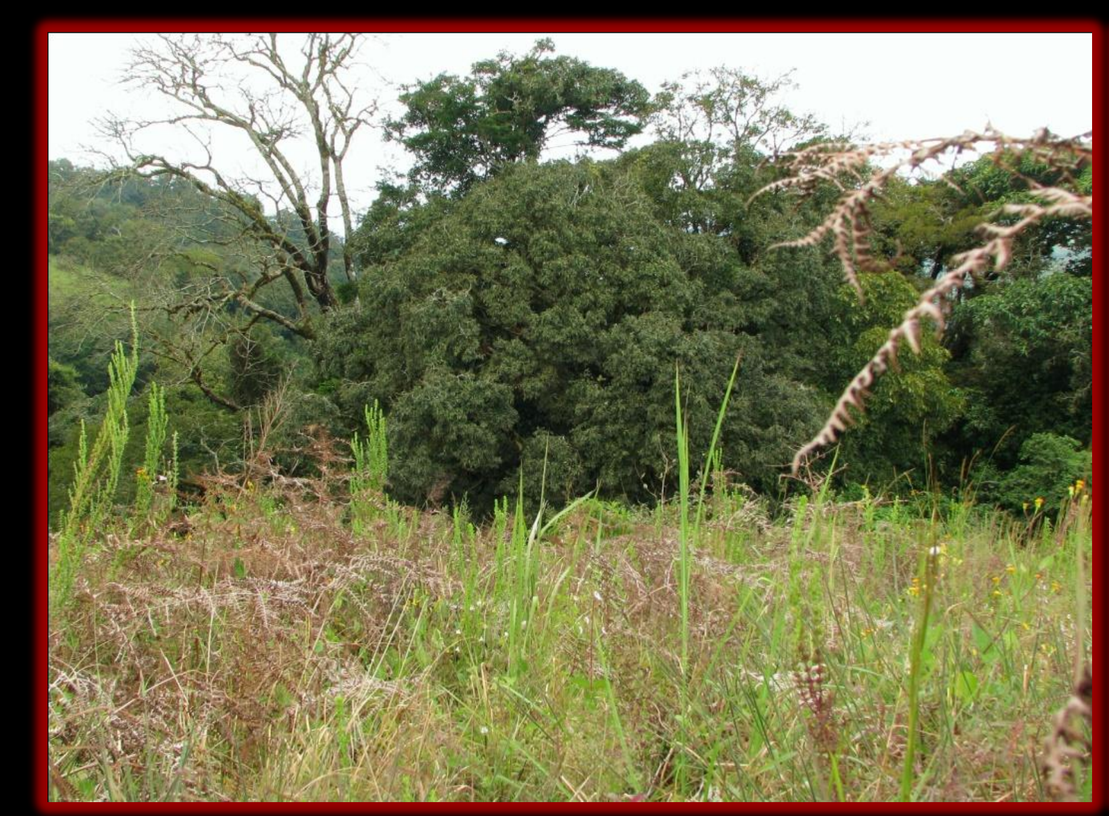

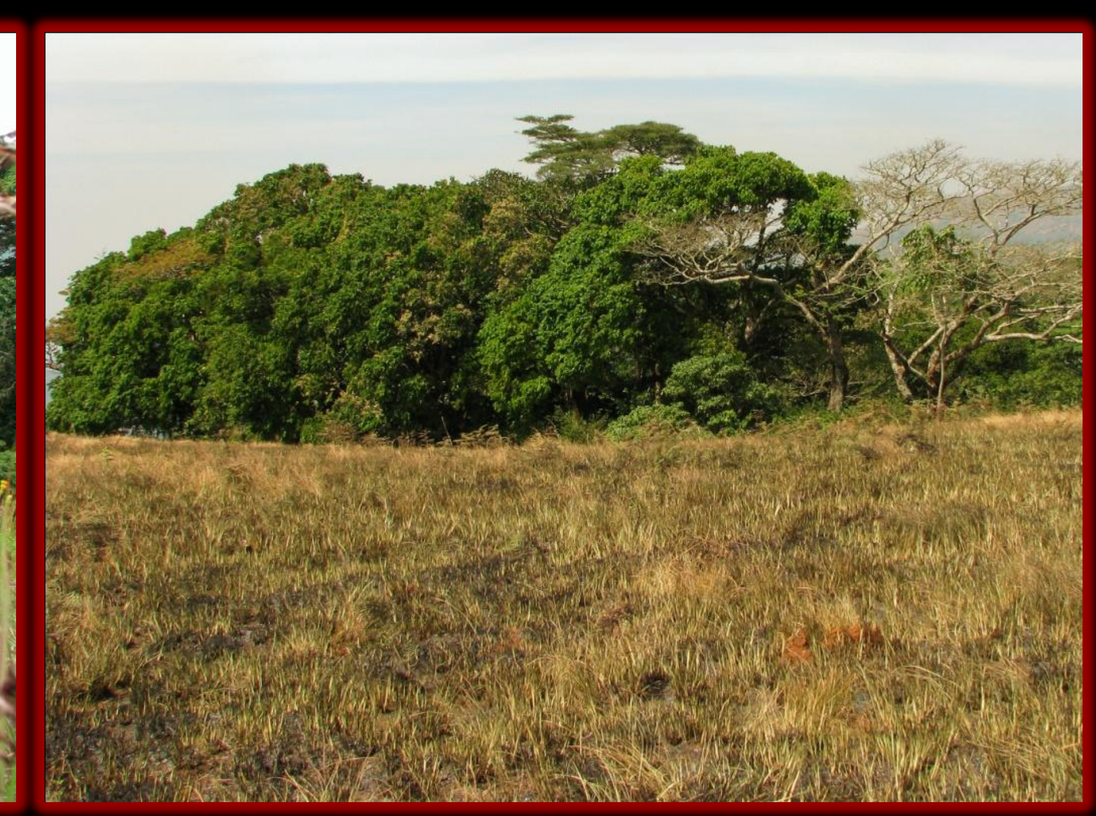

Using pig dung baited pitfall traps, dung beetles were sampled at distances of -160, -80, -40, -20, -10, -5, 0, 5, 10, 20, 40, 80, and 160 metres from the forest edge. Traps were at least 50m apart to maintain independence.

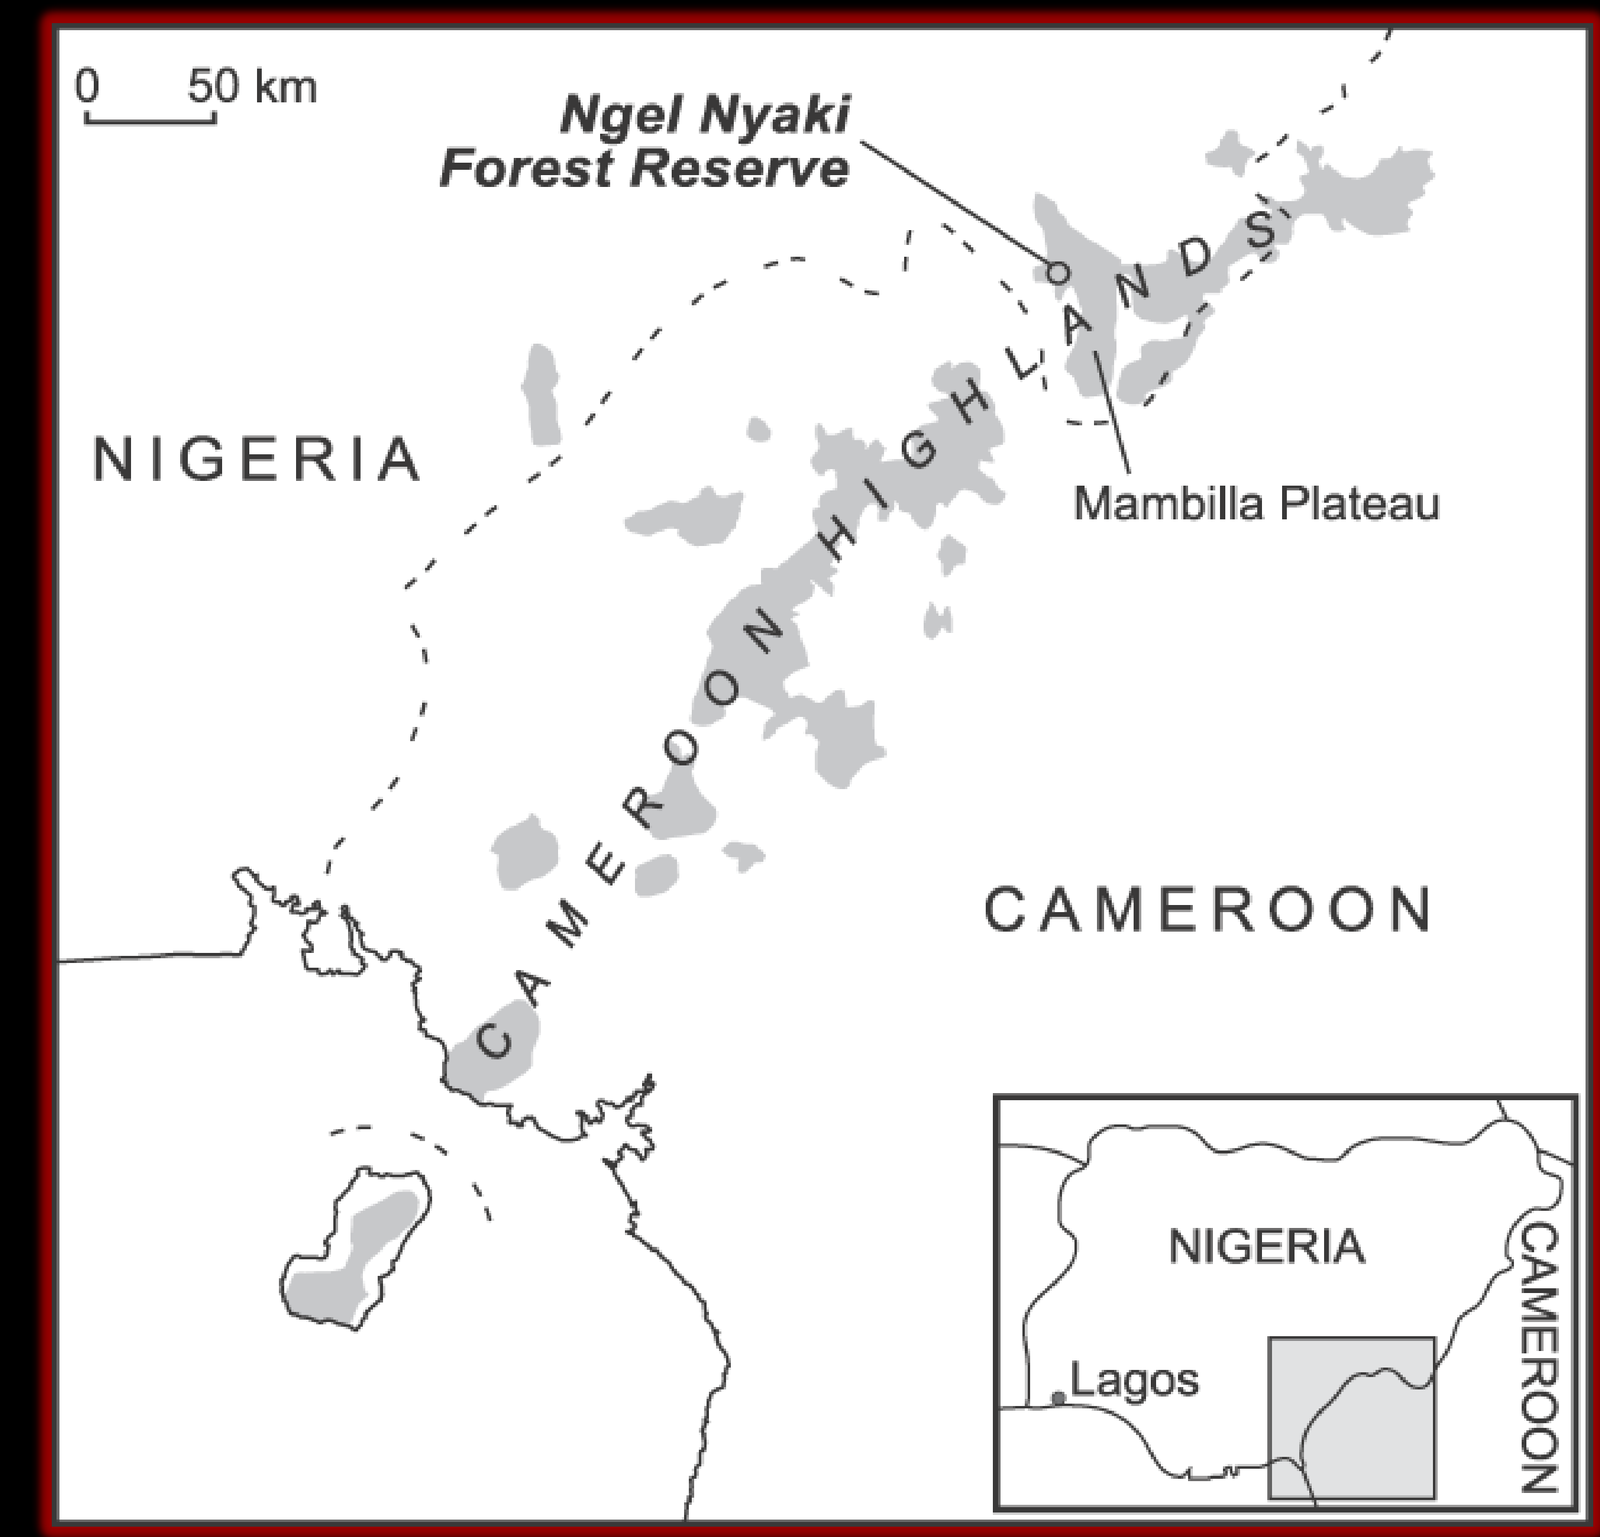
0 50 km
Ngel Nyaki Forest Reserve
NIGERIA
CAMEROON HIGHLANDS
Mambilla Plateau
CAMEROON
NIGERIA
Lagos
CAMEROON

Location of Ngel Nyaki forest reserve on the Mambilla Plateau, Nigeria. The inset rectangle shows the location of the main figure[2].

Trait-Dependent Responses

The extinction proneness of a species can often be determined by their traits (response traits). Dung beetles have been shown to exhibit trait-dependent responses to anthropogenic disturbance[3], but there is still no evidence of such responses across habitat edge gradients. Furthermore, anthropogenic disturbance may impact the ecosystem functions carried out by dung beetles (effect traits).

By analysing trait-dependent responses of dung beetles across the forest edge, we may then be able to determine possible correlations between these response and effect traits.

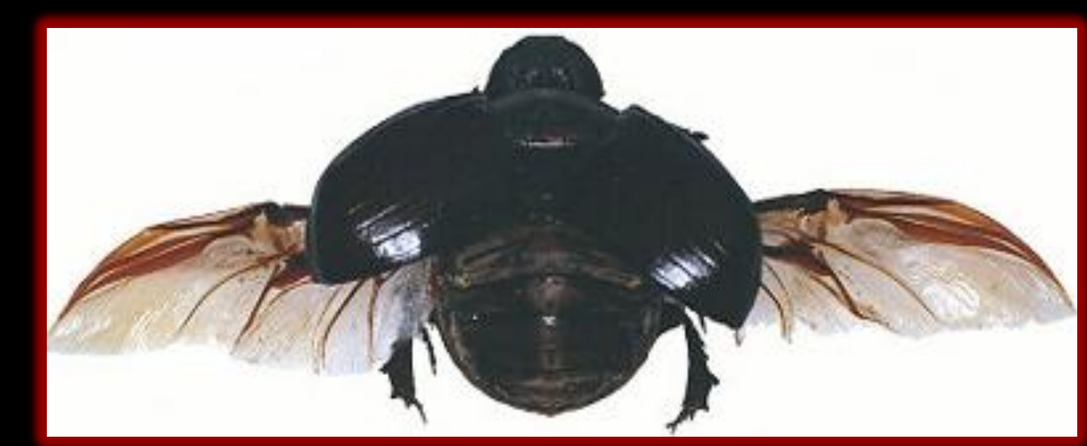

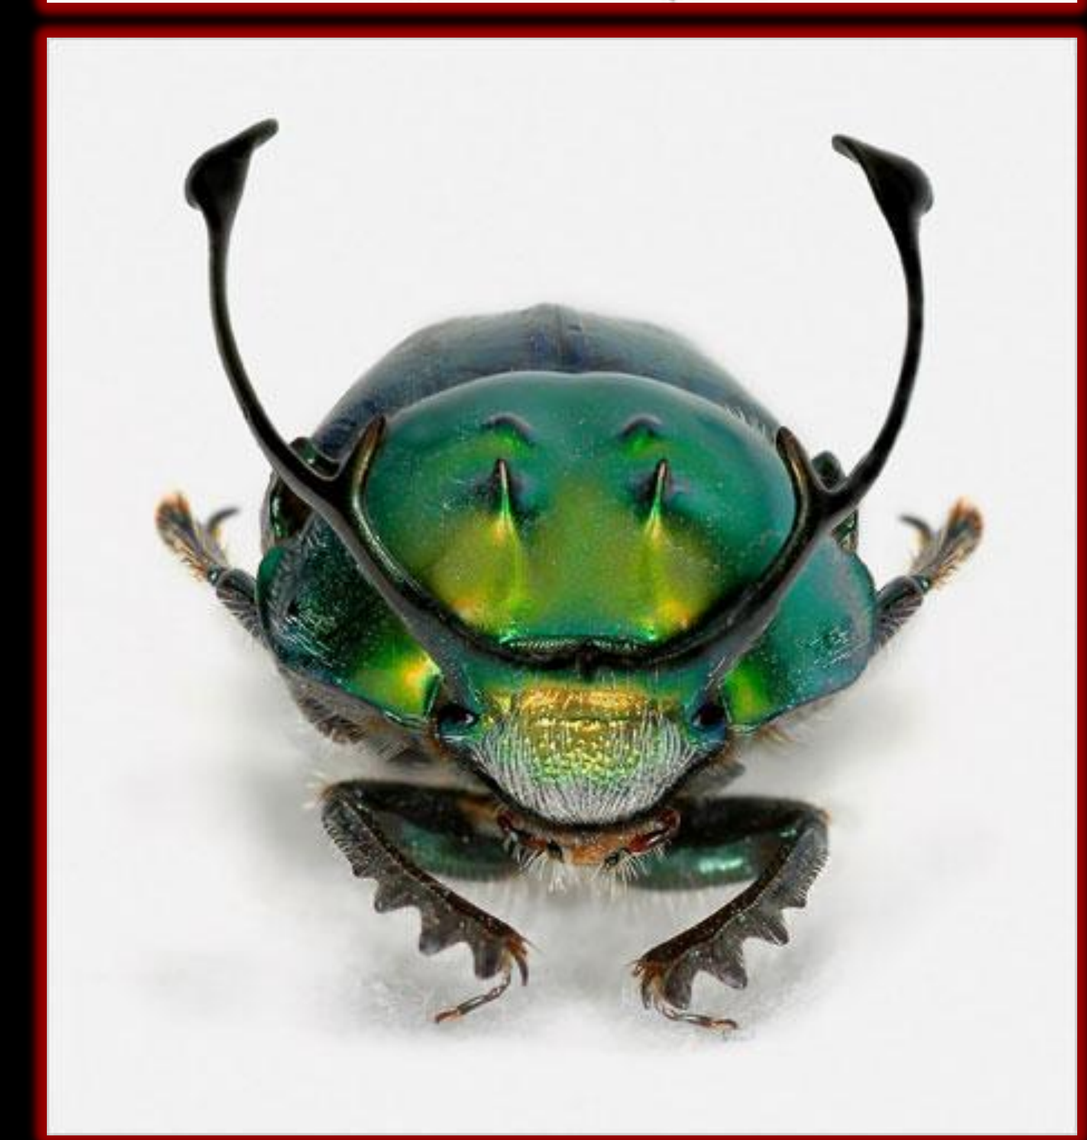

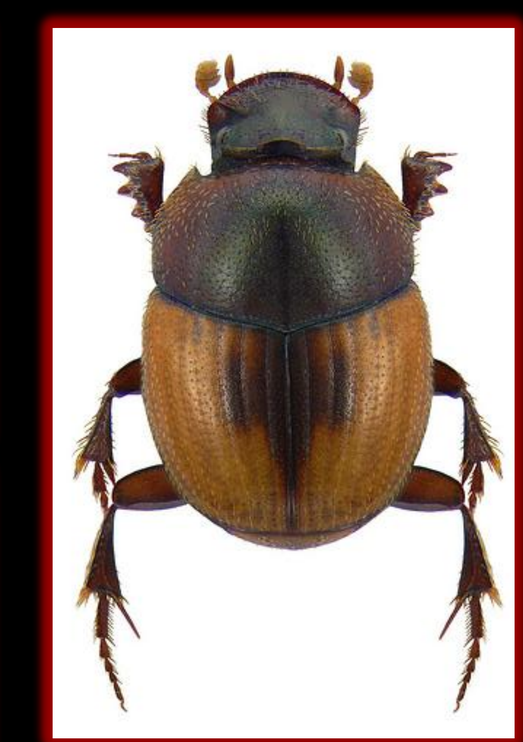

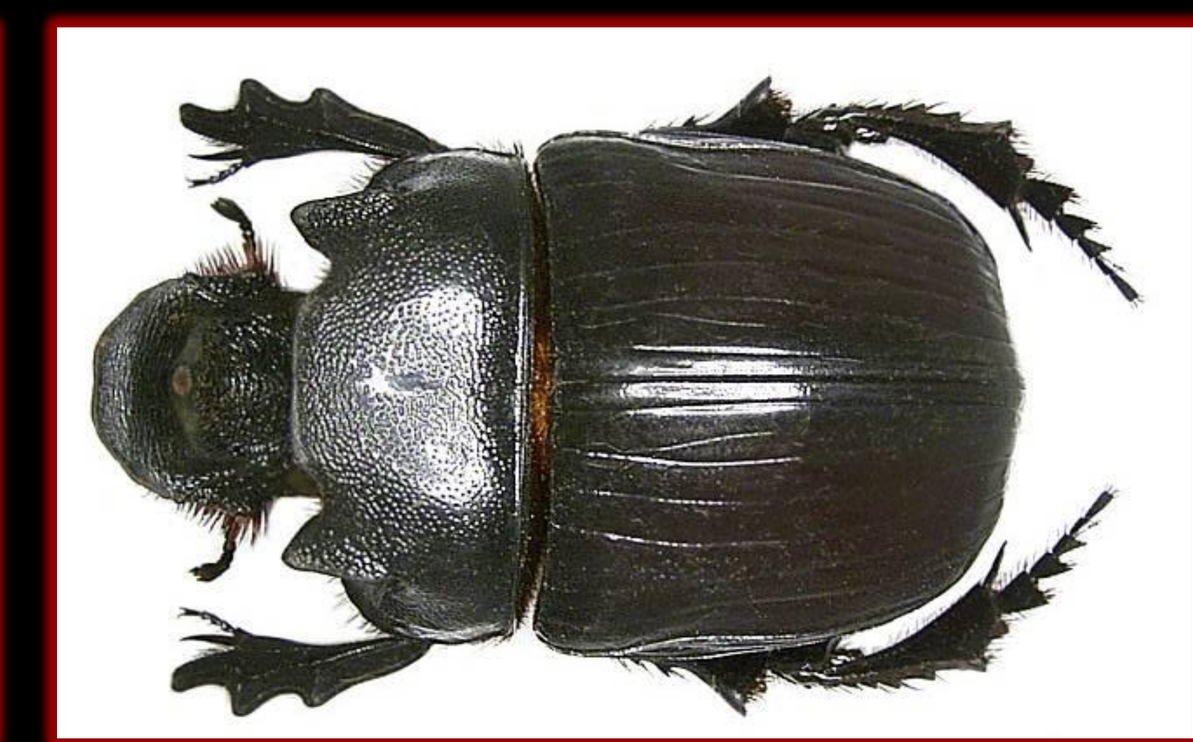

Body mass, body length and width, and wing area will be measured. Body size in dung beetles may be a strong determinant of susceptibility to disturbance. It may also account for much of the variation in ecosystem functioning as larger beetles remove more dung. We will also calculate wing loading (body mass/wing area) which will serve as a surrogate for dispersal ability, as this trait may also be important in determining species' susceptibility.

Yet To Be Dung...

Results show that dung beetle communities respond strongly to habitat edges. To what degree these responses occur will be identified as other parameters are analysed, such as species richness and species traits. The most important factor determining edge response in dung beetle community structure and ecosystem function, appears to be the degree of edge protection from livestock and fire encroachment. A major goal is to quantify these effects on secondary seed dispersal performed by dung beetles. This would provide insight into the possible ramifications of edge effects in forest maintenance and restoration through the effects on dung beetle communities. This research may potentially identify adverse correlations in community responses by comparing trait-mediated susceptibility with ecosystem functions performed by dung beetles.

Acknowledgements

I thank the Nigerian Montane Forest Project for providing the opportunity to carry out research at Ngel Nyaki forest reserve. I am especially grateful to Idirisa Musa for his assistance in the field and to Kristy Udy for her ongoing suggestions and support throughout this project.

Literature Cited

1. Ewers, R. M., and R. K. Didham. 2008. Pervasive impact of large-scale edge effects on a beetle community. Proceedings of the National Academy of Sciences of the United States of America **105:5426-5429.**
2. Beck, J., and H. Chapman. 2008. A population estimate of the Endangered chimpanzee Pan troglodytes vellerosus in a Nigerian montane forest: implications for conservation. Oryx 42:448-451.
3. Larsen, T. H., N. M. Williams, and C. Kremen. 2005. Extinction order and altered community structure rapidly disrupt ecosystem functioning. Ecology Letters 8:538-547.