Neonatal Glycaemic Control: Model Validation and *In-Silico* Virtual Patient Trials

A. Le Compte, A. Lynn, J.G. Chase, G.M. Shaw, G. Russell, A. Blakemore, D. Lee, X.W. Wong, J. Lin, C.E. Hann

INTRODUCTION

Premature, low-birth-weight infants in the neonatal intensive care unit (NICU) can lose blood glucose homeostasis due to immaturity of endogenous regulatory systems and the stress of their condition.

Typical treatment relies on glucose restriction before insulin administration due to fear of hypoglycaemia. However, glucose restriction may adversely affect the infant's nutrition status.

A model of the fundamental glucose regulatory dynamics can provide insight about the metabolic state of the patient. *In-silico* virtual trials can be used to design optimal insulin therapy regimes for this vulnerable patient group.

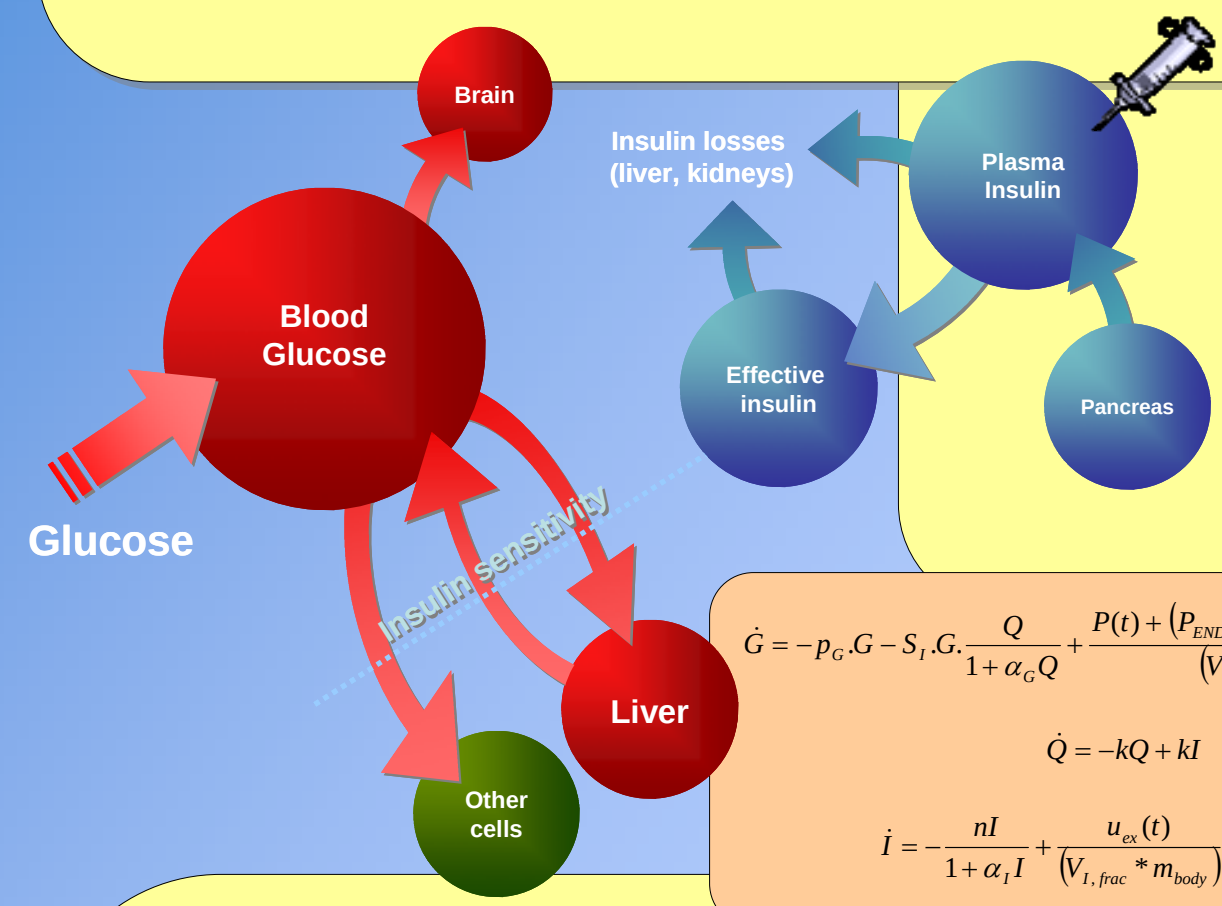
Brain
Insulin losses (liver, kidneys)
Plasma Insulin
Blood Glucose
Effective insulin
Pancreas
Glucose
Insulin sensitivity
Liver
Other cells

$$\dot{G} = -p_G.G - S_I.G.\frac{Q}{1+\alpha_G Q} + \frac{P(t) + (P_{END} * m_{body}) - (CNS * m_{brain})}{(V_{G,frac}(t) * m_{body})}$$

$$\dot{Q} = -kQ + kI$$

$$\dot{I} = -\frac{nI}{1+\alpha_I I} + \frac{u_{ex}(t)}{(V_{I,frac} * m_{body})} + e^{-(k_I u_{ex}(t))} I_B$$

MODELS AND METHODS

An adult critical care metabolic system model is adapted to the unique physiological case of the neonate. Time dependent volumes of distribution, insulin clearance and central nervous system uptake are significantly different in the neonate compared to adult.

Integral-based methods identify time-varying insulin sensitivity. Optimal non-insulin mediated glucose uptake and endogenous glucose production parameters selected through grid-search.

Retrospective clinical for N=25 cases contained 1091 glucose measurements over 3567 total patient hours plus all insulin and nutritional infusion data.

Retrospective patient cohort details:

No. patients	Total hours	Total BG measurements	Avg. gestational age at birth	Age at start of fitting (days)	Avg. weight at birth
25	12.7	8.1	23	< 1	600g

Model-prediction based bisection algorithm used to optimise insulin dosage during simulated trials. Typical sensor measurement errors of 2.5%-7% included to assess robustness of algorithm. BG measurement frequency optimised to reduce nursing workload and stress on limited infant blood volume.

RESULTS: MODEL VALIDATION

The identified model had a median absolute percentage error of 2.4% [IQR: 1.0%-4.9%]. Median absolute prediction errors at 1, 2 and 4 hours were 5.2% [IQR: 2.5%-10.4%], 9.5% [IQR: 4.5%-18.5%] and 13.7% [IQR:6.3%-27.8%] respectively.

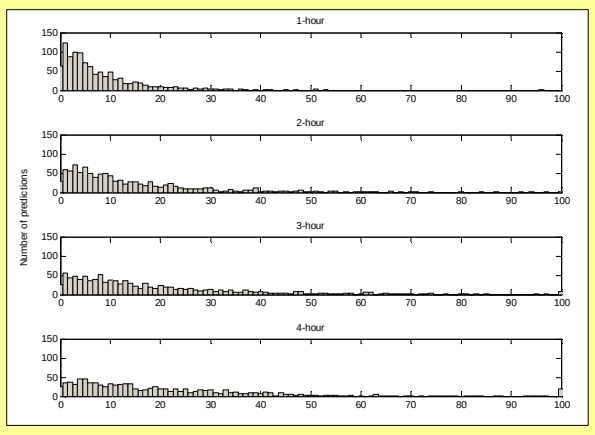
1-hour
2-hour
3-hour
4-hour
Number of predictions

Absolute percentage BG prediction error for 1, 2, 3 and 4 hour prediction intervals.

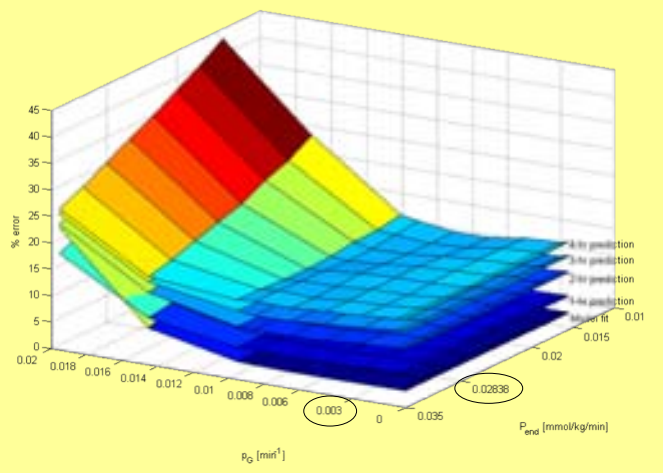
% error
0.003
0.02838
p_G [min^{-1}]
P_{end} [mmol/kg/min]

Fitting and prediction errors for several combinations of model parameters p_G and P_{END}. The optimal combination of (p_G, P_{END}) are circled.

Non-insulin mediated uptake p_G and endogenous glucose production P_{END} optimised in simulation. Model fitting and prediction performance was robust to (p_G, P_{END}) selection. Optimal endogenous glucose production agrees with tracer-measured studies in literature.

Identified insulin sensitivity shows significant per-patient variation, agreeing with literature conclusions of great heterogeneity in neonatal response to insulin and glucose infusions.

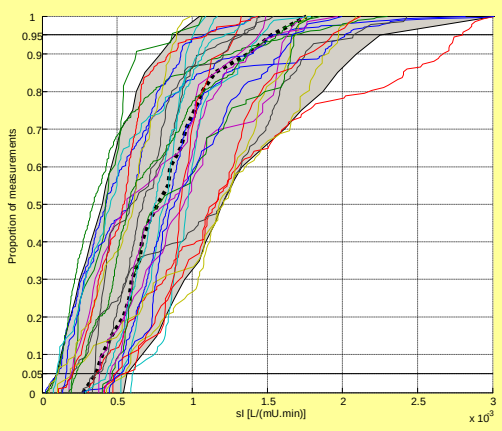
Proportion of measurements
sI [L/(mU.min)]

Cumulative distribution of insulin sensitivity (S_I) per-patient. The dashed line and shaded area represent the median and 5%-95% distribution of S_I.

RESULTS: VIRTUAL TRIALS

Virtual trials modulated insulin infusions utilising model-based predictions on 25 patient profiles.

Two-hourly measurement regime simulation achieved 76.1% of measurements within target 4-7 mmol/L band, compared to 30.9% for retrospective records. Median BG reduced 25% from 8.0 to 6.0 mmol/L.

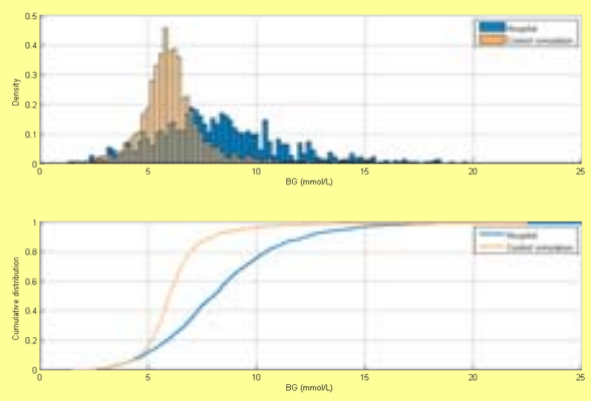

Cumulative distribution (bottom panel) and histogram (top panel) of BG measurements for retrospective and simulated results.

More frequent BG measurement and intervention achieved greater glycaemic control performance.

Simulated BG control performance for 1-4 hour BG measurement and model-based intervention periods compared to hospital control.

	Hospital	1 hour	2 hour	3 hour	4 hour
Num. of measurements	1091	3555	1771	1175	879
BG median [mmol/L]	8.0	6.1	6.0	6.0	6.1
BG IQR [mmol/L]	[6.3 - 9.9]	[5.6 - 6.6]	[5.4 - 6.8]	[5.3 - 7.0]	[5.2 - 7.2]
% of BG within 4-7 mmol/L	31%	82%	76%	69%	65%

Optimal measurement frequency balances control performance with clinical requirements.

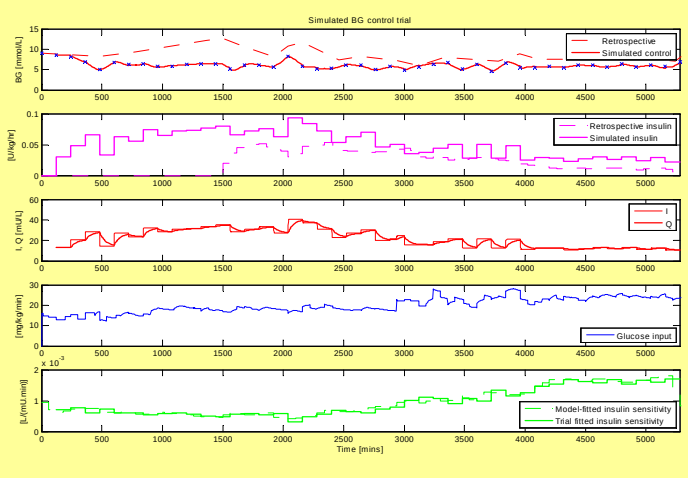
Simulated BG control trial
Retrospective
Simulated control
Retrospective insulin
Simulated insulin
I
Q
Glucose input
Model-fitted insulin sensitivity
Trial fitted insulin sensitivity
Time [mins]

Simulated trial results for Patient #9 comparing retrospective hospital control records (dashed lines) with model-based controller (solid lines).

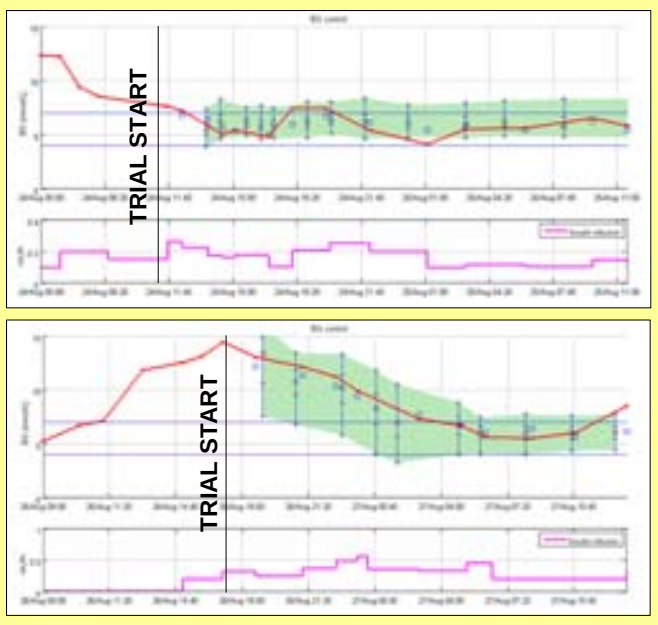
TRIAL START
TRIAL START

Clinical BG results for pilot trial patients 2 (top panel) and 3 (bottom panel). Green areas denote stochastic model 5%-95% BG prediction confidence interval.

Proof-of-concept clinical trials performed in Christchurch Neonatal Unit. Insulin infusion rates modulated by model-based controller for up to 24 hours per patient.

Canterbury District Health Board
Te Poari Hauora ō Waitaha

UC BIOMEDICAL ENGINEERING

UC UNIVERSITY OF CANTERBURY
Te Whare Wānanga o Waitaha
CHRISTCHURCH NEW ZEALAND