A Simple, High-Resolution Model-Based Insulin Sensitivity Test - Analysis, Benchmark and Clinical Pilot

Thomas Lotz[1], J Geoffrey Chase[1], Kirsten A McAuley[2], Geoffrey M Shaw[3], Juliet Berkeley[3] and Jim I Mann[2]

[1]Centre for Bioengineering, University of Canterbury, Christchurch, New Zealand
[2]Edgar National Centre for Diabetes Research, Dunedin, New Zealand
[3]Dept of Intensive Care Medicine, Christchurch Hospital, Christchurch, New Zealand

UNIVERSITY of OTAGO Te Whare Wānanga o Otāgo | UC BIOENGINEERING | UC UNIVERSITY OF CANTERBURY Te Whare Wānanga o Waitaha CHRISTCHURCH NEW ZEALAND

1.0 Introduction

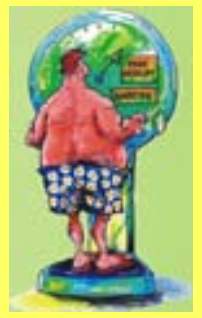

Type 2 Diabetes affects more than **230 million people** worldwide and is responsible for **3.5 million deaths** annually [1]. About **215 000 New Zealanders** are estimated to have type 2 Diabetes and an equal number are insulin resistant, prior to developing the disease [2]. Costs for the NZ public health system: **$540 million** in 2006, expected to rise over **$1.8 billion by 2021** [2]. Similar numbers and scales hold worldwide [1].

Early diagnosis of insulin resistance can reduce the burden of further complications and save lives and cost. Current diagnostic methods are too expensive and complicated to be useful in widespread population screening (euglycaemic clamp, IVGTT), or not accurate enough to give more than a high/low result (HOMA, OGTT).

Insulin resistance rises up to 10 years prior to diagnosis

2.0 Test design

Provide complete diagnostic information about:
- **Insulin sensitivity**
- **β-cell function**

Designed to measure the <u>same</u> metabolic effect as the gold standard euglycaemic hyperinsulinaemic clamp.

<u>Design goals</u>:
1.) Simple, low intensity, **2.)** high accuracy, **3.)** low cost, and **4.)** physiologically dosed (and thus measured)

<u>The protocol</u>:

1.Low dose bolus of glucose (10g) and insulin (1U)
- ➡ Reduce endogenous regulatory effects
- ➡ Improve clinical practicability and safety

2. 30 min timed sampling of glucose, insulin and C-peptide

3. Fit metabolic models
- ➡ Only transient data from 10 - 30 minutes post bolus
- ➡ Identification requires only few samples

4. Determine insulin sensitivity from model parameter S_I

5. Determine β-cell function from C-peptide data

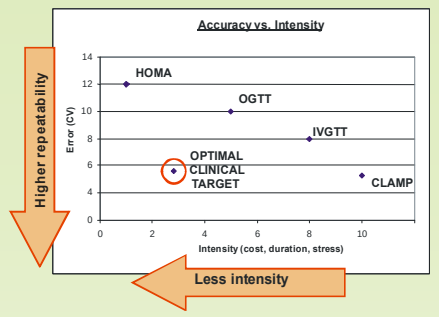
Accuracy vs. Intensity
Higher repeatability
Error (CV)
HOMA
OGTT
IVGTT
OPTIMAL CLINICAL TARGET
CLAMP
Intensity (cost, duration, stress)
Less intensity

1.Glucose + insulin bolus

2. 30 min timed blood sampling

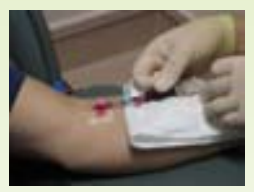

3. Fit metabolic models

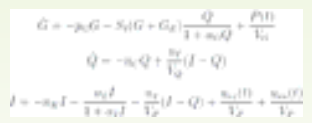

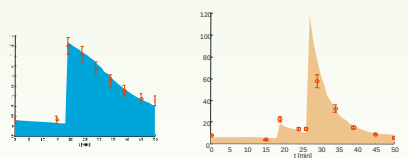

4. Determine insulin sensitivity

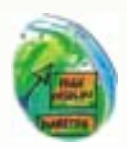

5. Determine b-cell function

•Basal secretion
•1st phase secretion

3.0 Diagnostic outcome

Test outcome on 3 exemplary subjects spanning the range from NGT to Type 2 Diabetes

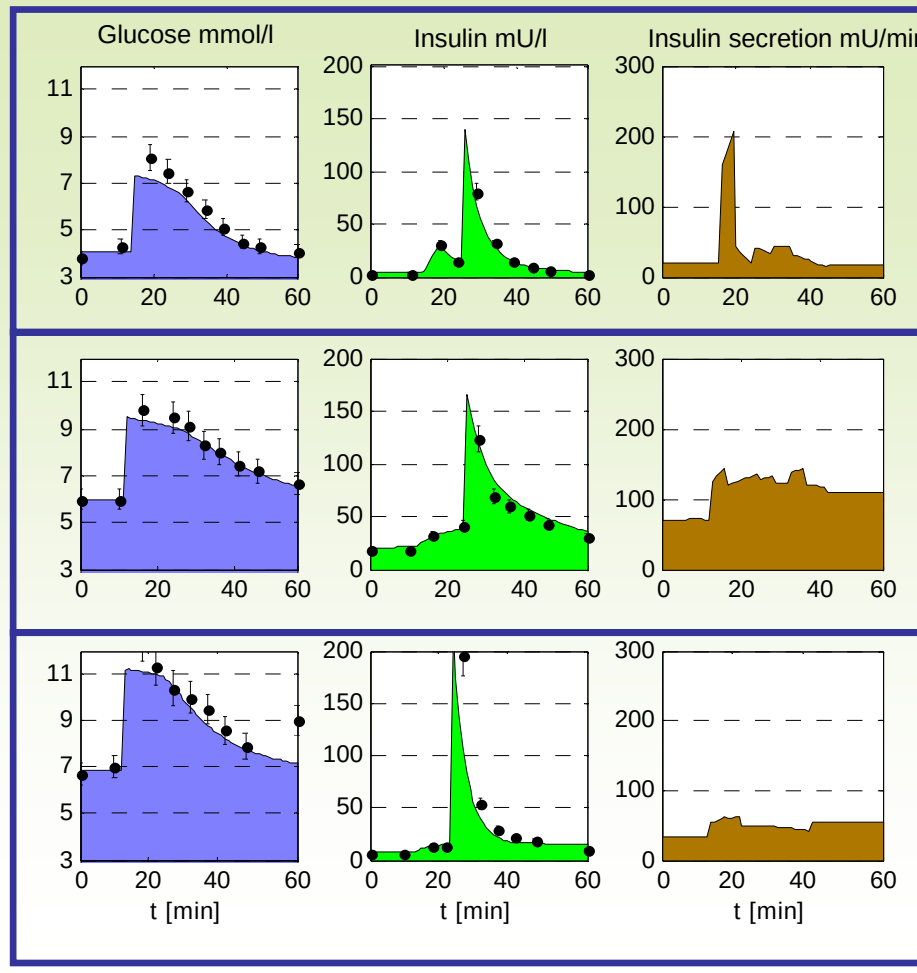
Glucose mmol/l
Insulin mU/l
Insulin secretion mU/min
t [min]
t [min]
t [min]

<u>Normal Glucose Tolerant (NGT)</u>
- •High insulin sensitivity (11.7 x 10^{-4} mU/l/min)
- •Healthy 1st phase insulin response
- •Rapid decay of glucose concentration

<u>Impaired Fasting Glucose (IFG)</u>
- •Low insulin sensitivity (3.2 x 10-4 mU/l/min)
- •Impaired 1st phase insulin response
- •Elevated insulin secretion throughout
- •Slow decay of glucose concentration

<u>Type 2 diabetes</u>
- •Low insulin sensitivity (6.7 x 10-4 mU/l/min)
- •Insignificant 1st phase insulin response
- •Significant b-cell damage
- •Elevated glucose concentrations

4.0 Performance

1. <u>Model validation on euglycemic clamp data</u> [3]

a) Correlation clamp ***ISI*** vs. model S_I **r=0.97**

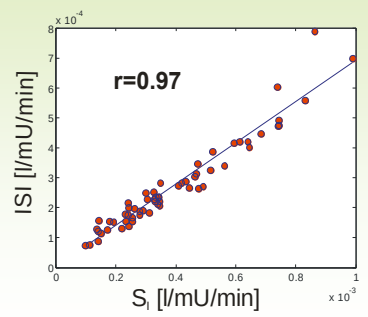
r=0.97
ISI [l/mU/min]
S_I [l/mU/min]

b) Change in insulin sensitivity in intervention study by McAuley et al. [4]

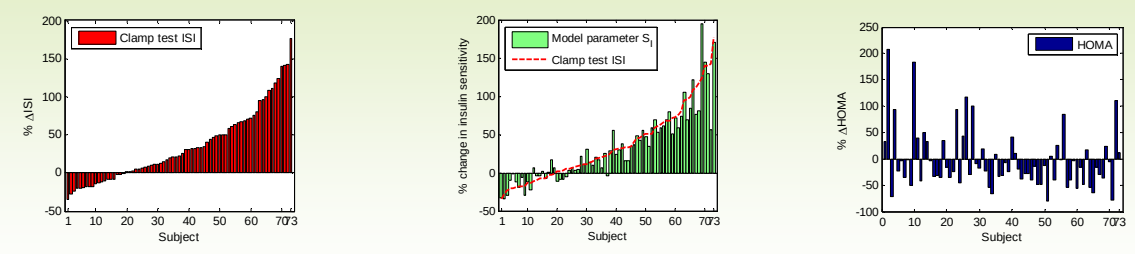
Clamp test ISI
% ΔISI
Subject
Model parameter S_I
Clamp test ISI
% change in insulin sensitivity
Subject
HOMA
% ΔHOMA
Subject

Clamp (dynamic, steady state) | **Our model (dynamic, transient state)** | **HOMA (static fasting metric)**

2. <u>Clinical Pilot study</u>:
17 subjects, 43 tests

<u>Part 1:</u> Effect of dose on outcome
- Low dose: 5g glucose / 0.5U insulin
- Medium dose: 10g glucose / 1U insulin
- High dose: 20g glucose / 2U insulin

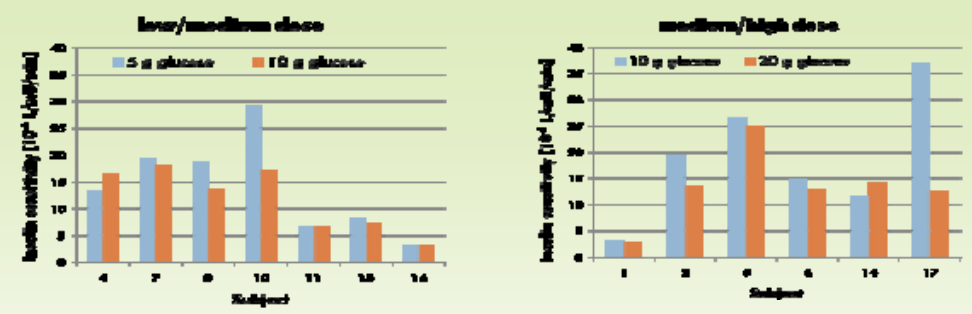

Difference not statistically significant (P=0.5, P=0.52)

<u>Part 2:</u> Repeatability
- Same dose on same individual on a different day

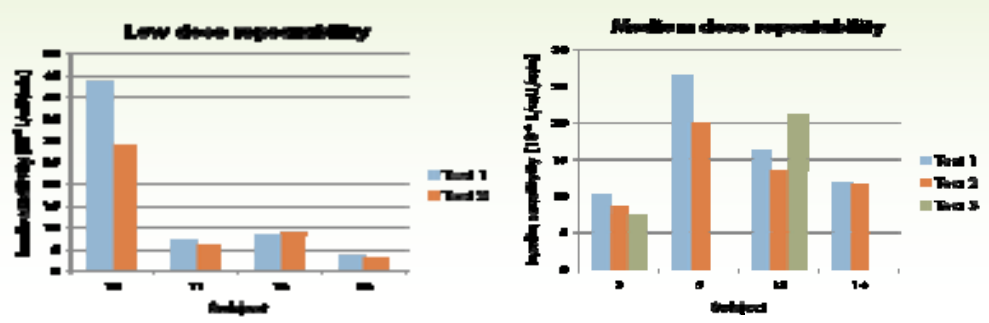

Error 6.0% (MSD 4.9)
Range 1.7% - 24.7%

[1] International Diabetes Federation (www.idf.org)
[2] PWC for Diabetes New Zealand (2007), "Type 2 Diabetes – Outcome model update"
[3] Lotz T, Chase JG, et al. (2006), "Transient and steady state euglycemic clamp validation of a model for glycemic control & insulin sensitivity testing", DT&T 8(3)
[4] McAuley K, et al. (2002), "Intensive Lifestyle Changes Are Necessary to Improve Insulin Sensitivity", Diabetes Care 25(3)
[5] McAuley K, et al. (2007), "Point: Homeostasis Model Assessment ", Diabetes Care 30(9)